Optimal Drug Product Presentation and Container Closure Selection for AAV-Based Genomic Medicines

Madhura Som, Diego Vargas, Varna Kanapuram, Benson Gikanga, Katelyn Esposto, Michelle Jones, Sandeep Yadav
Sangamo Therapeutics Inc.

Poster # 547

Introduction

The majority of AAV-based commercial drug products and clinical drug candidates are stored at frozen conditions due to the potential impact on stability associated with long term 2-8°C storage. While conventional container closure systems (CCS) such as glass vials offer unique benefits like low product adsorption and impermeability to moisture and gas exchange, the risk of glass breakage with frozen AAV storage necessitates the assessment and implementation of alternative container systems. Polymeric vials are a great option to address these challenges, however, they may not provide chemical durability needed to ensure the drug product remains stable and efficacious throughout the drug product life cycle. In addition, the physical durability of the inner surface of vials is crucial and needs to be assessed for any corrosion or delamination. This study evaluates the compatibility of AAV-based products and related manufacturing stresses in commercially available polymeric vials.

Drug Product (DP) Container Compatibility: Background

The aim of this poster is to illustrate how polymeric and hybrid vials, due to their mechanical strength, are a good choice for frozen AAV DP formulations. Specifically, hybrid vials have features of "glass-like" coated surface deposited over a cyclic olefin polymer (COP) body that prevent gas permeation through the walls.

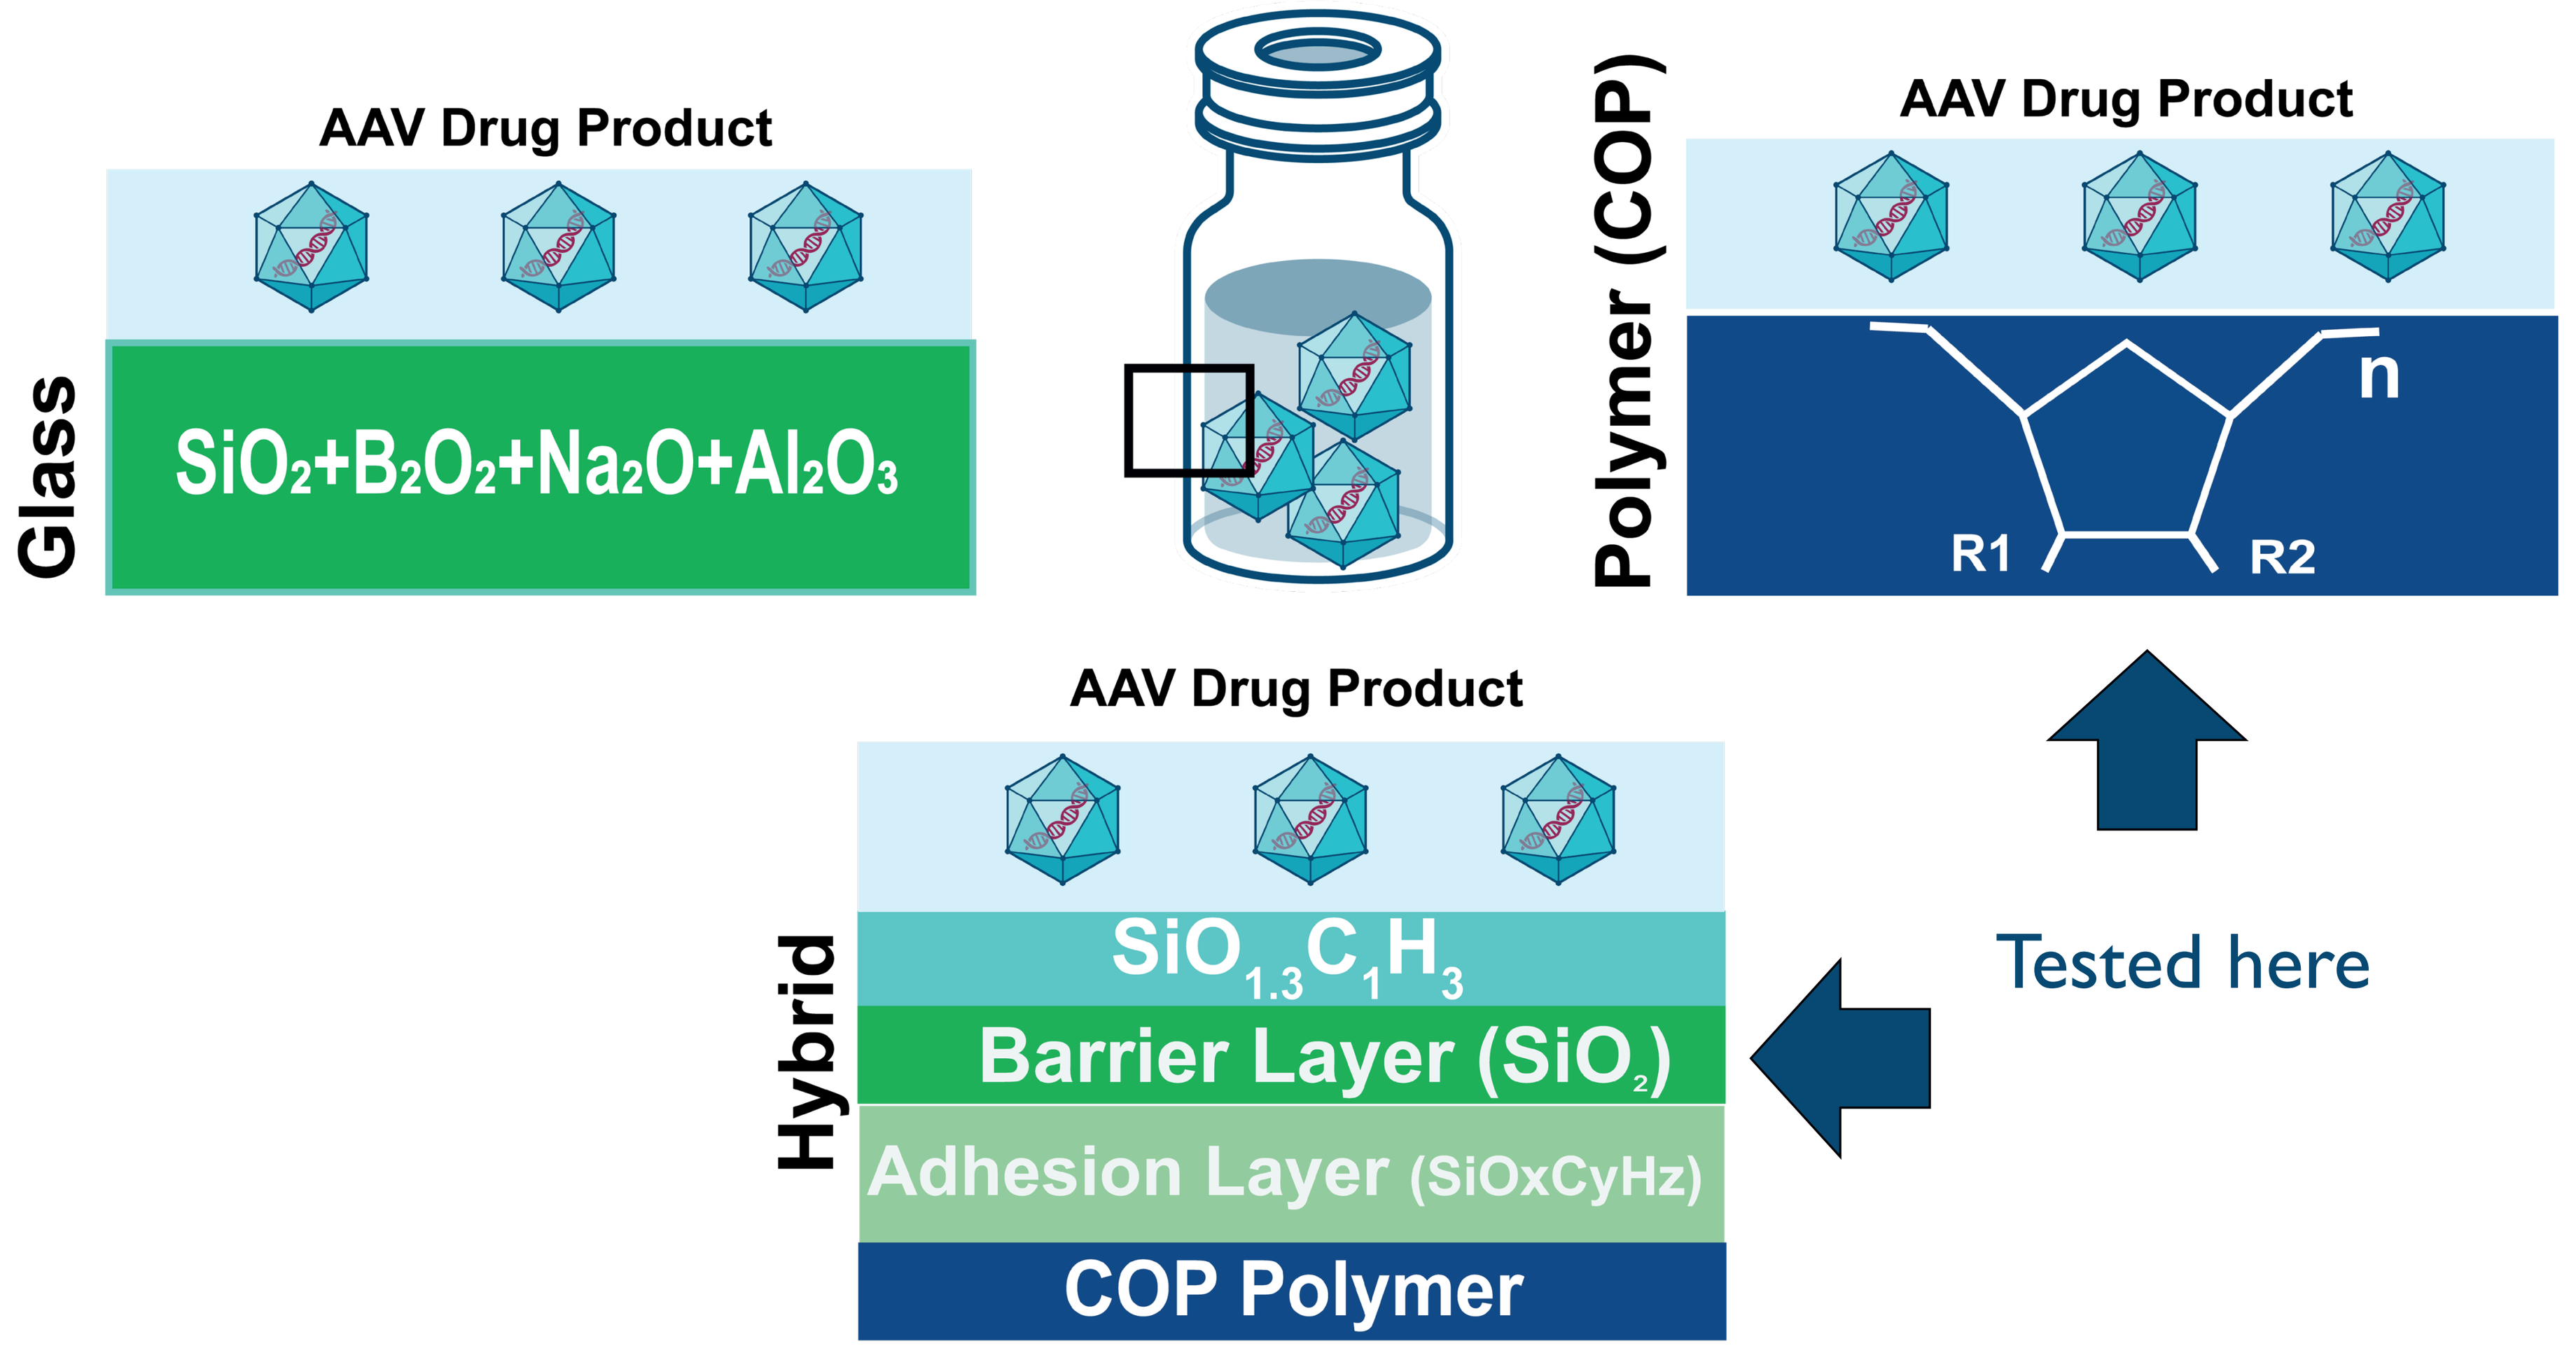
AAV Drug Product
Glass
SiO_2+B_2O_2+Na_2O+Al_2O_3
Polymer (COP)
AAV Drug Product
n
R1
R2
AAV Drug Product
Hybrid
$SiO_{1.3}C_1H_3$
Barrier Layer (SiO_2)
Adhesion Layer (SiOxCyHz)
COP Polymer
Tested here

Figure 1. DP vial Material of Construction (MoC) types: Glass, Polymer (e.g., Cyclic Polyolefin) and Hybrid (polymer with a protective layer of glass). In this study, we compare the performance of COP and Hybrid Vials with an AAV-based DP formulation.

SvP analysis with BMI shows differences between COP and Hybrid Vials

COP and hybrid 2mL vials evaluated for compatibility with a frozen AAV-based DP in a phosphate buffered formulation intended for frozen long-term storage. CQAs Evaluated: Vector genome titer, Capsid titer, Free DNA and High Molecular Weight Species up to 90 days head-to-head comparison between polymeric vs hybrid vials (Figure 2).

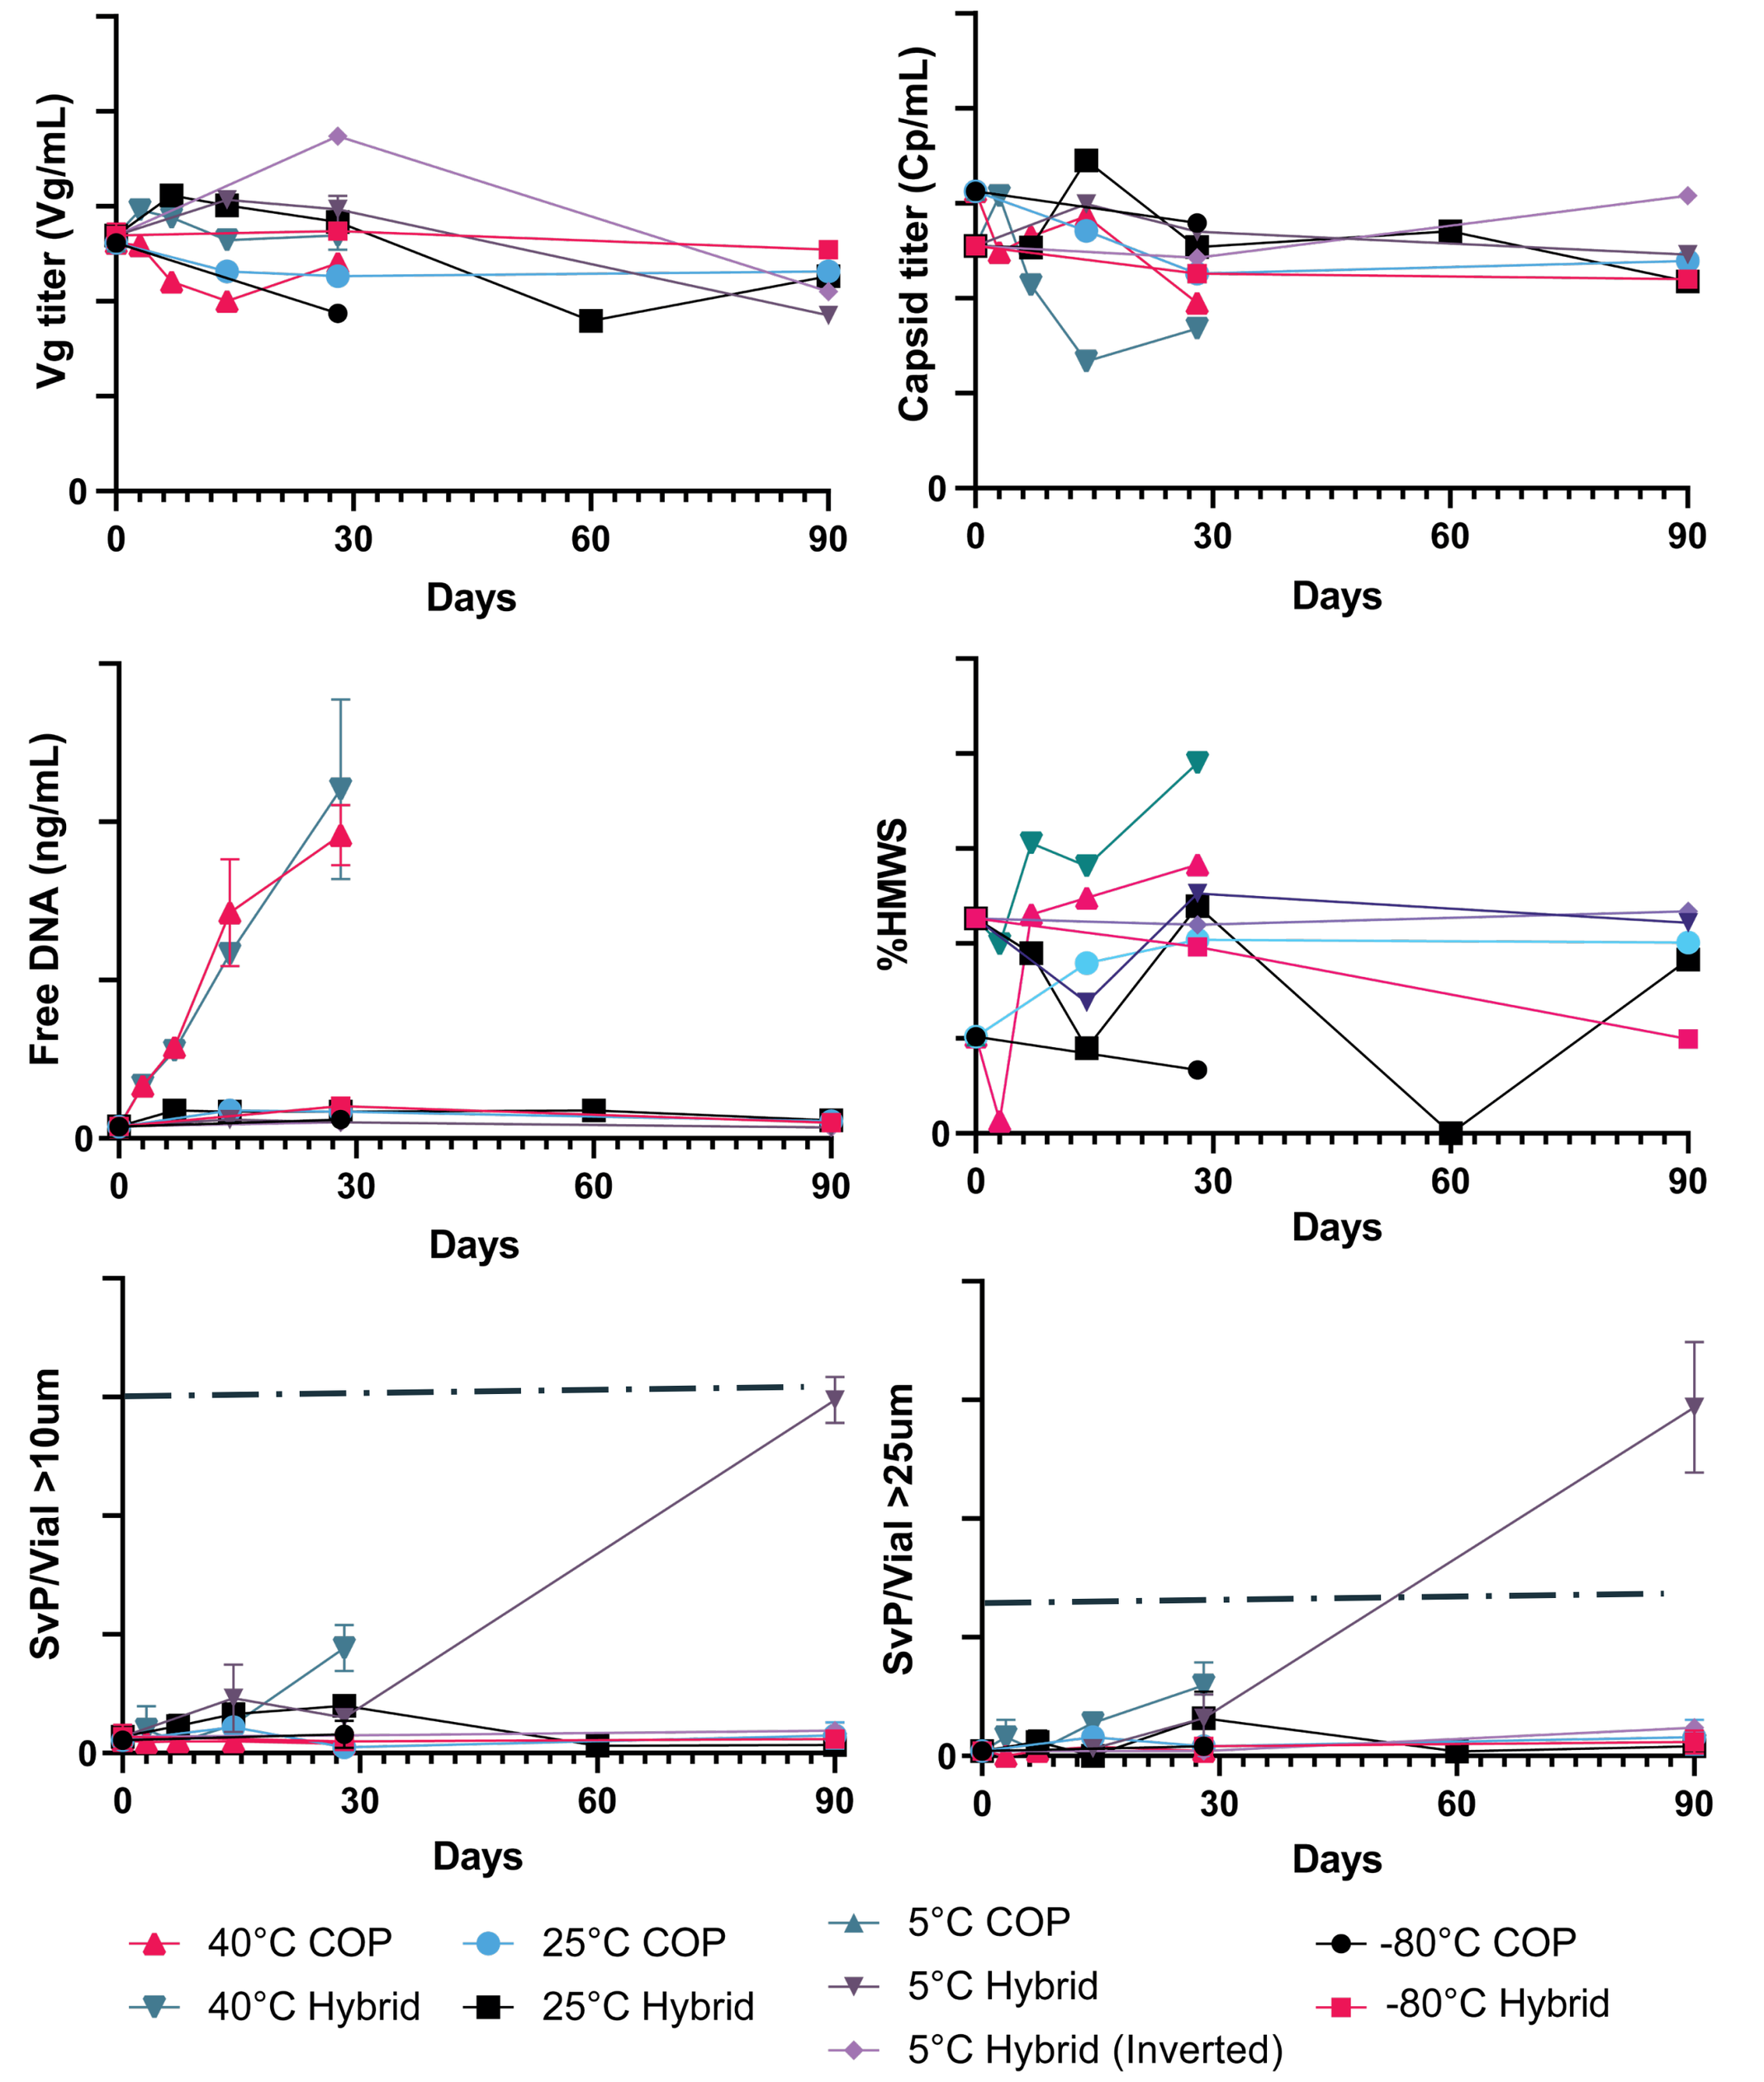
Vg titer (Vg/mL)
Capsid titer (Cp/mL)
Free DNA (ng/mL)
%HMWS
SvP/Vial >10um
SvP/Vial >25um
Days
0
30
60
90
40°C COP
40°C Hybrid
25°C COP
25°C Hybrid
5°C COP
5°C Hybrid
5°C Hybrid (Inverted)
-80°C COP
-80°C Hybrid

Figure 2. Vg titer (Top, left), Capsid titer (Top, right), Free DNA (Middle, left), %HMWS (Middle, right) and Subvisible particulates (SvP) by Background Membrane Imaging (Bottom, left ≥10 μm and Bottom, right ≥25 μm for AAV DP in COP and Hybrid Vials for up to 90 days. Dotted lines in SvP graphs show the limits/container per USP <787> or <788>.

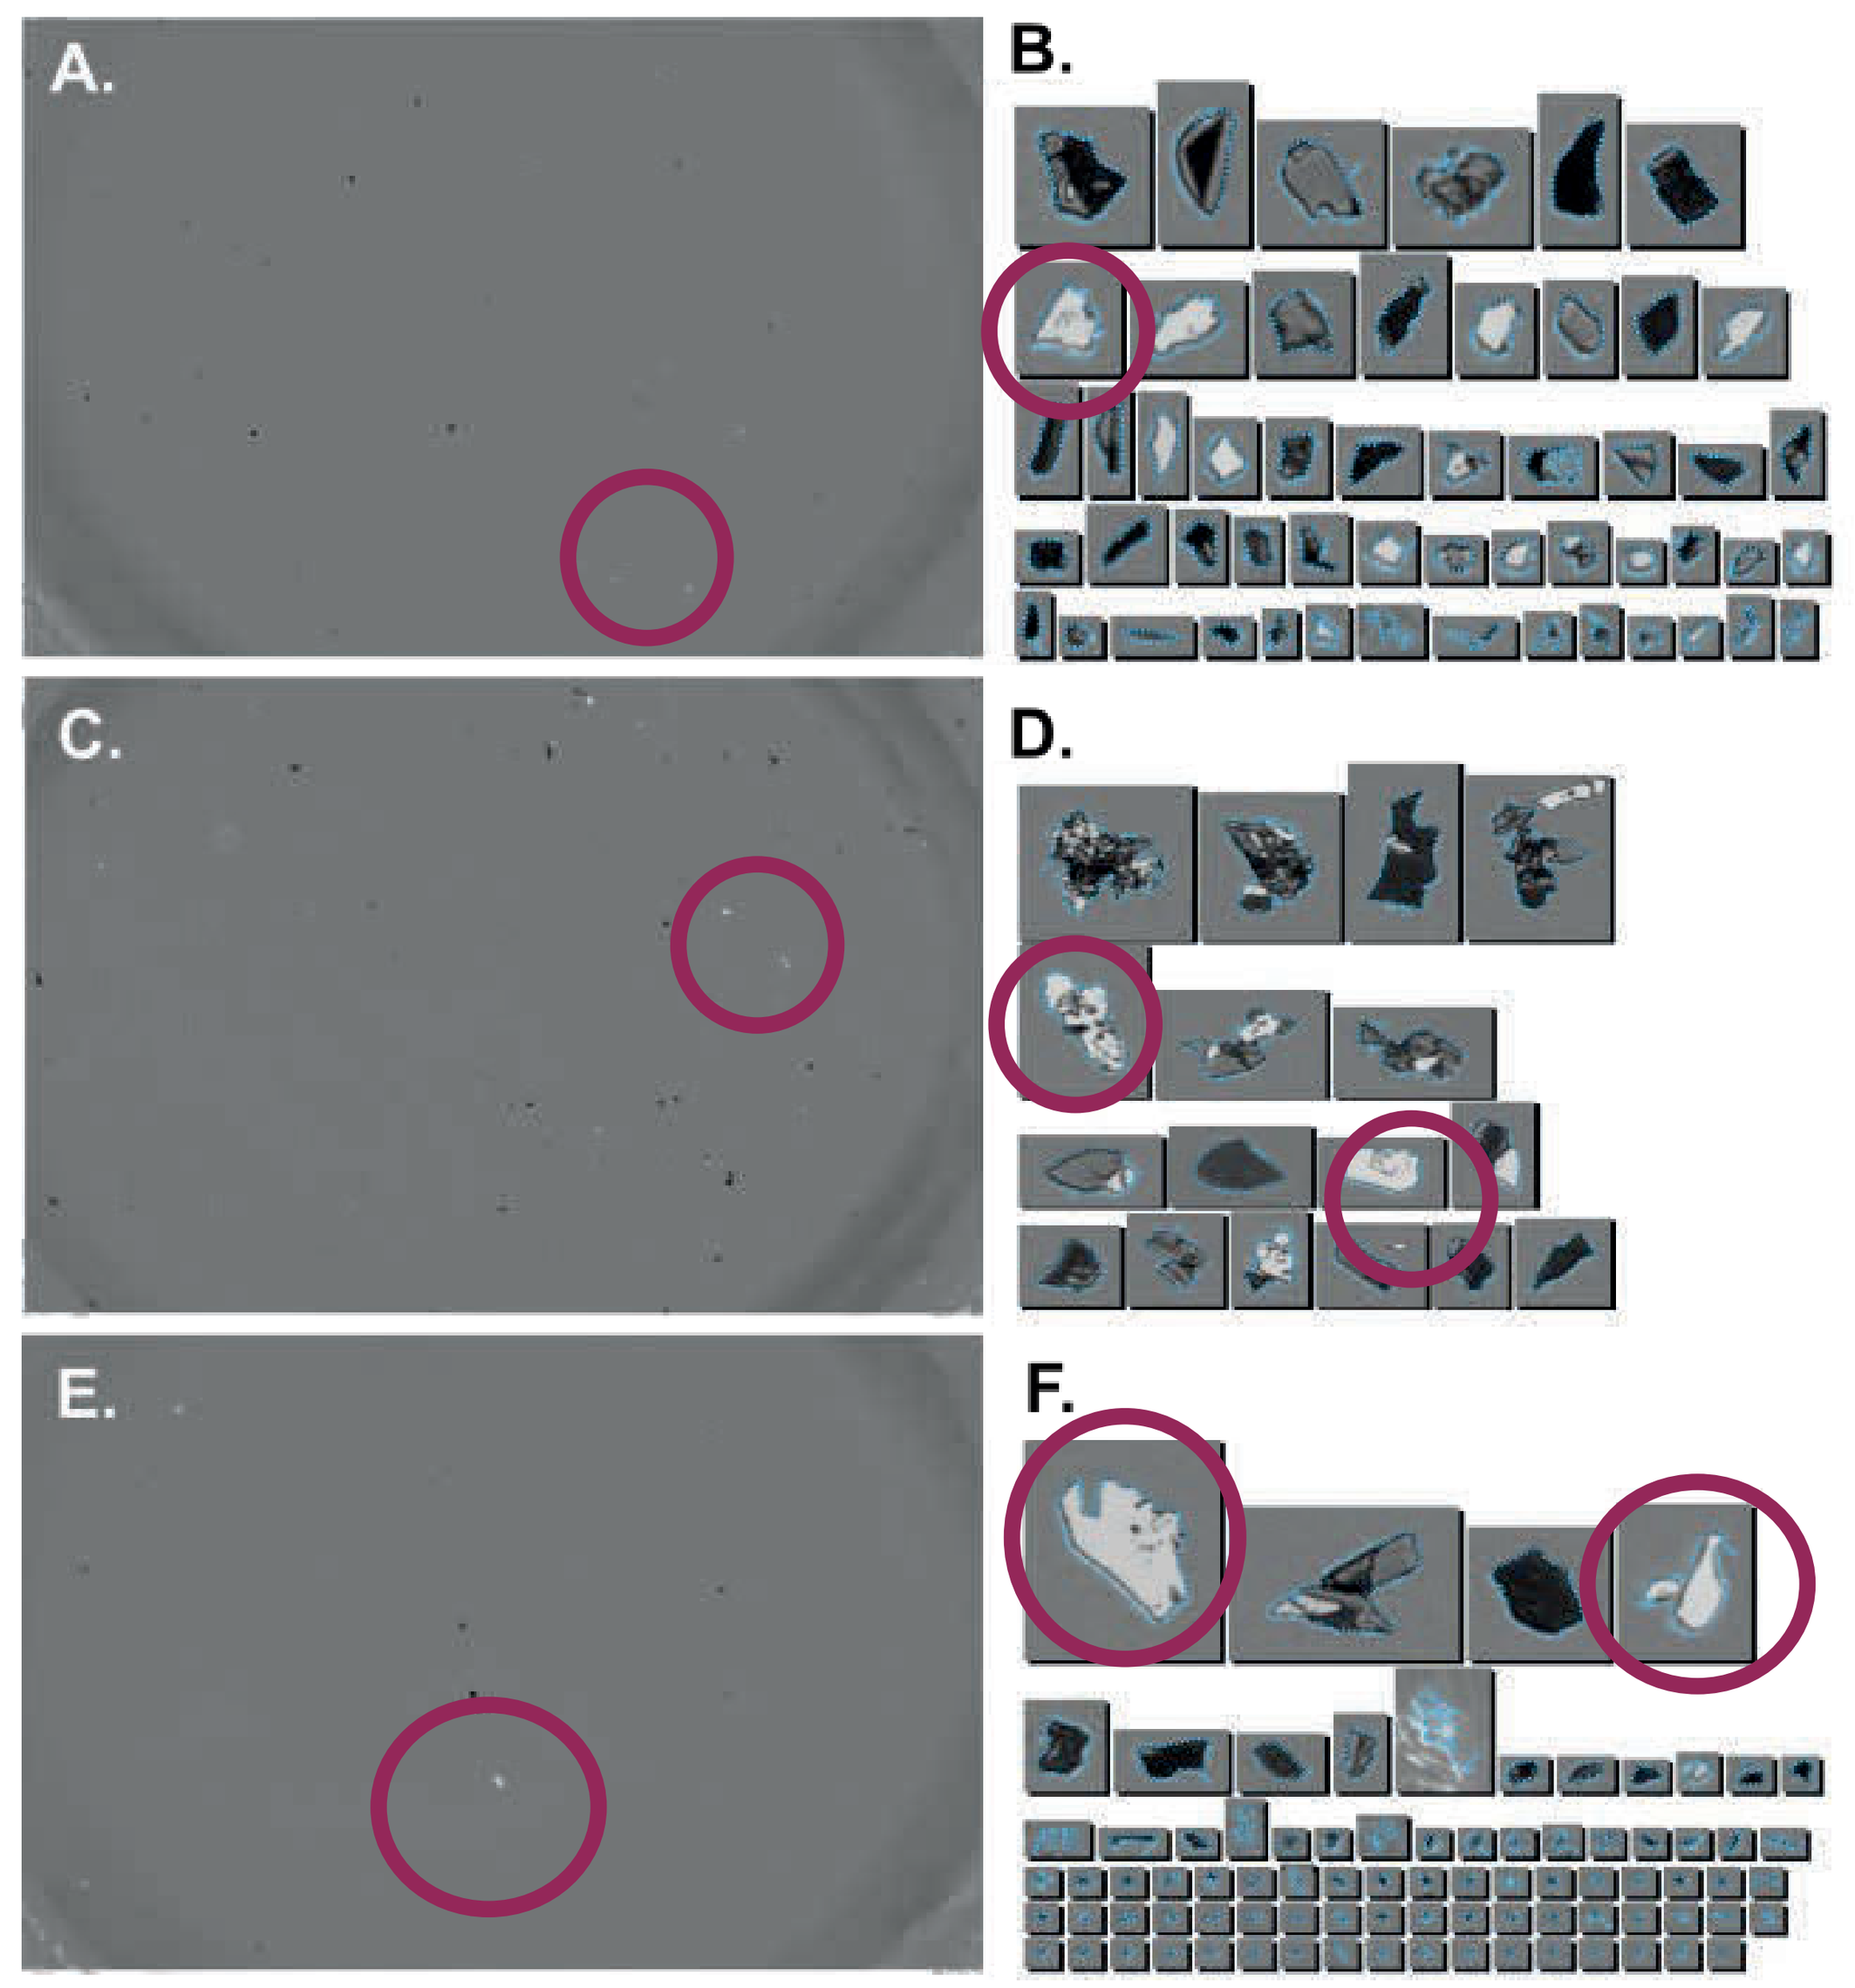
A.
B.
C.
D.
E.
F.

Figure 3. Background membrane imaging (BMI) based SvP particulate analysis: plate (A, C, E) and particle morphology images (B, D, F) showing subvisible particulates deposited from AAV DP stored in Hybrid Vials at (A, B) 40°C for 28 days, (C, D) 5°C for 90 days, (E, F) 7x freeze/thaw cycles. Red circles indicate white flakes that showed up for AAV DP stored in hybrid vials but not in COP vials.

Subvisible particle (SvP) counts characterized by a background membrane imaging technique (BMI, Figure 3) showed higher SvP for DP stored in hybrid vials. BMI showed differences in SvP morphologies i.e., white flakes detected with the hybrid vials but not from the COP vials. Further investigation was performed to determine the chemical composition of the particulates using scanning electron microscopy (SEM) combined with Energy Dispersive Spectroscopy (EDS).

SEM with EDS confirmed that the particles are from the surface coating on Hybrid Vials

Stereo-microscopy and scanning electron microscopy (SEM) performed on the hybrid vials w/ AAV DP in 5°C (inverted configuration) and 25°C conditions showed:

- Circumferential scattering band indicating possible coating defects (neck region)
- Flake like particles (~20-80μ) in SEM that showed the presence of Si, O and C elements with EDS, consistent with the coating chemistry of the hybrid vials

These observations suggest that the flakes are fragments of the coating material of the inner surface of the hybrid vials.

Figure 4. Stereo-microscopy images of the upper part of the vial body (A,B), SEM images (E,G) and EDS spectra (I) found in hybrid vials exposed to AAV-based DP at 25°C for 90 days. Stereo-microscopy images of the upper part of the vial body (C,D), SEM images (F,H) and EDS spectra (J) found in hybrid vials exposed to AAV DP at 5°C (inverted configuration) for 90 days. EDS spectra show the presence of Si, O, and C while Ag comes from the background substrate used for the imaging.

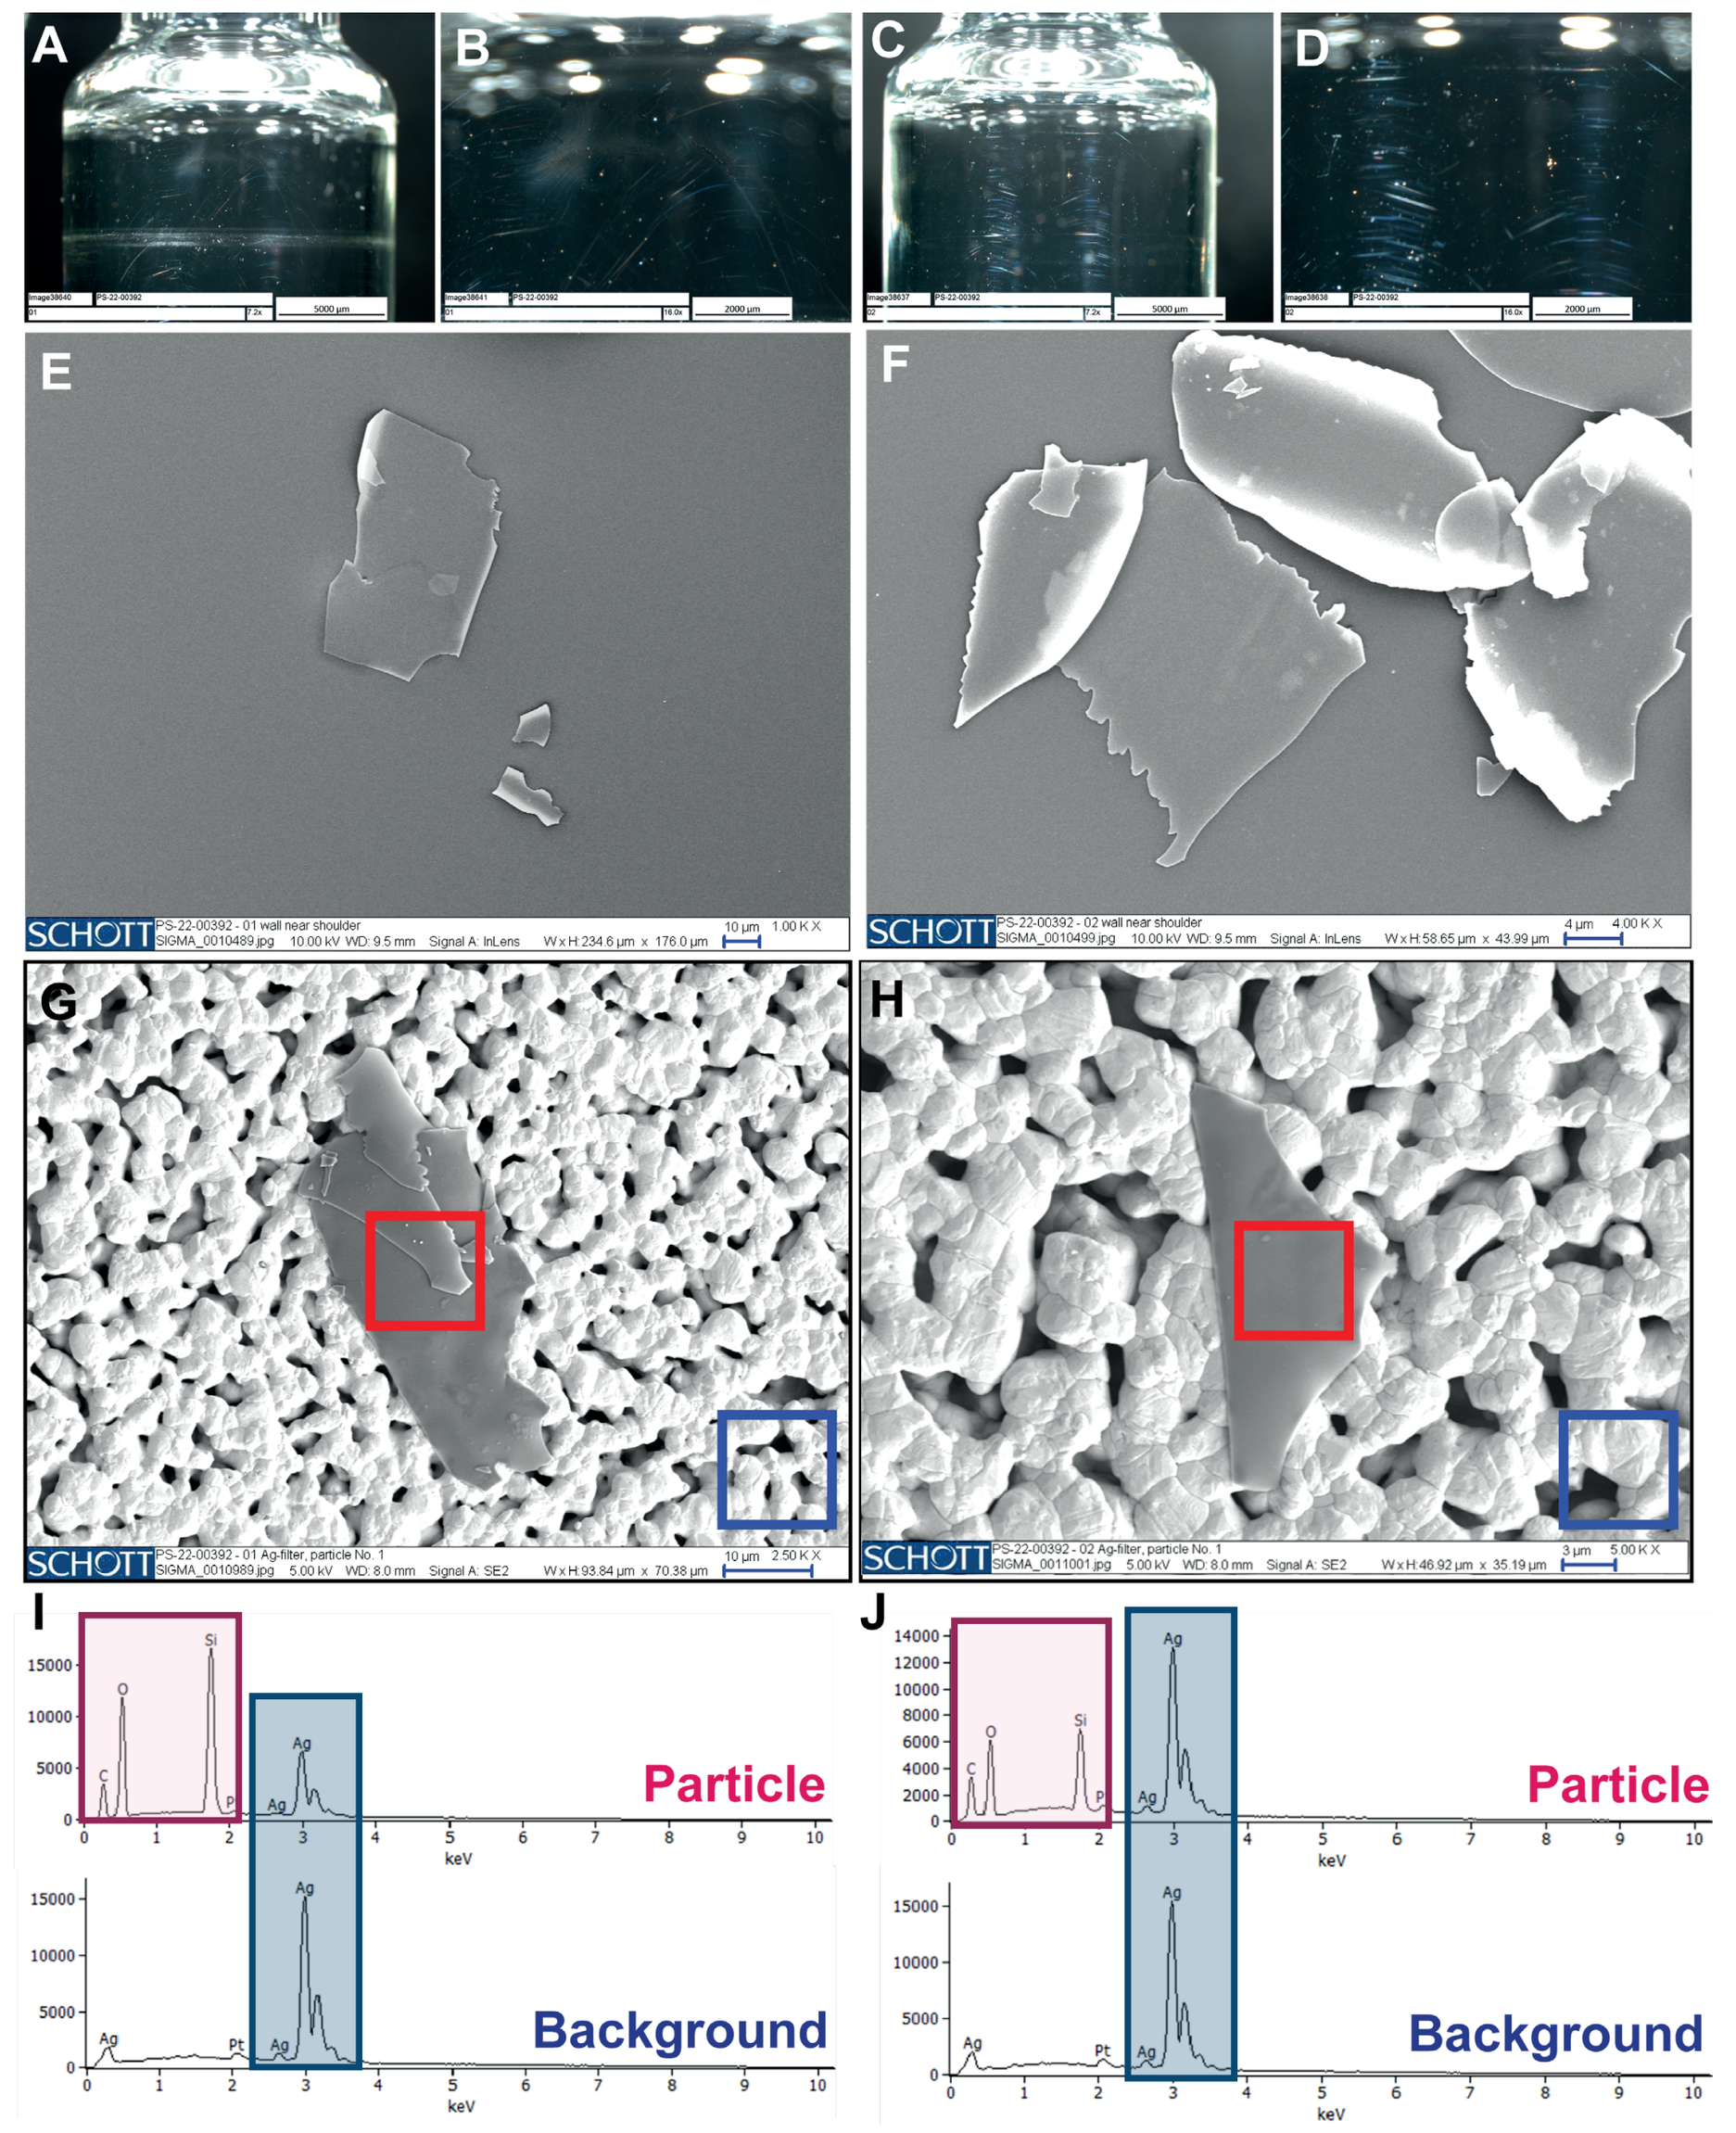
A
B
C
D
E
F
G
H
I
J
SCHOTT
Particle
Background
keV

**Vials tested were hybrid type and not manufactured by Schott

Summary & Conclusions

- Particulate testing with BMI, SEM and EDS showed presence of micron sized (~20-80μ) white flakes that seemed to be dislodged from the inner coating of the hybrid vials. No such effect was observed with COP vials.
- BMI and SEM confirmed the presence of these flakes are related to structural defect of the hybrid vials and possible delamination of surface coating.
- This work underscores the importance of optimal DP container selection and a holistic analysis of DP container compatibility for a given DP presentation.

Acknowledgements

We thank SCHOTT Pharma USA, Inc. for SEM, EDS & stereo-microscopic analysis of the hybrid vials & the Sangamo Formulation & Fill/Finish team for their support.

Presented at ASGCT 2024