Development of Blood-Brain Barrier Penetrant AAVs through Receptor-Targeted Capsid Engineering

Ankitha Nanjaraj*, Stephanye Frias*, Satria Sajuthi, Lori Andrews, Russell Darst, Stephen Wist, Yuri Bendaña, Hung Tran, Sarah Mueller, Bryan Zeitler, Amy M. Pooler, Matthew Tiffany, David S. Ojala
All authors are current or former employees of Sangamo Therapeutics, Inc., Richmond, CA.

Poster #985

Introduction

- The development of blood-brain barrier (BBB) penetrant AAV capsids has the potential to enable transformative genomic medicines for the treatment of neurological disorders.
- We have previously described SIFTER, a method for *in vivo* selection of AAV capsids that mediate improved central nervous system (CNS) delivery after administration into the vasculature or cerebrospinal fluid. *In vivo* SIFTER screens in non-human primates have yielded novel capsids that access the CNS after intravascular (STAC-BBB, Abstract #117) or cerebrospinal fluid (STAC-102 and STAC-103) administration.
- Here, we deployed a complementary library screening approach to engineer AAV capsids targeting receptors highly expressed in endothelial cells comprising the human blood-brain barrier.
- Multiplexed library screening was conducted for three distinct BBB receptors. For each receptor, library screens identified engineered capsids that exhibit enhanced transduction of cells that overexpress the receptor. A subset of these capsids also interact with macaque and mouse orthologs, suggesting the potential for cross-species CNS delivery.

Receptor-targeted capsid engineering approach

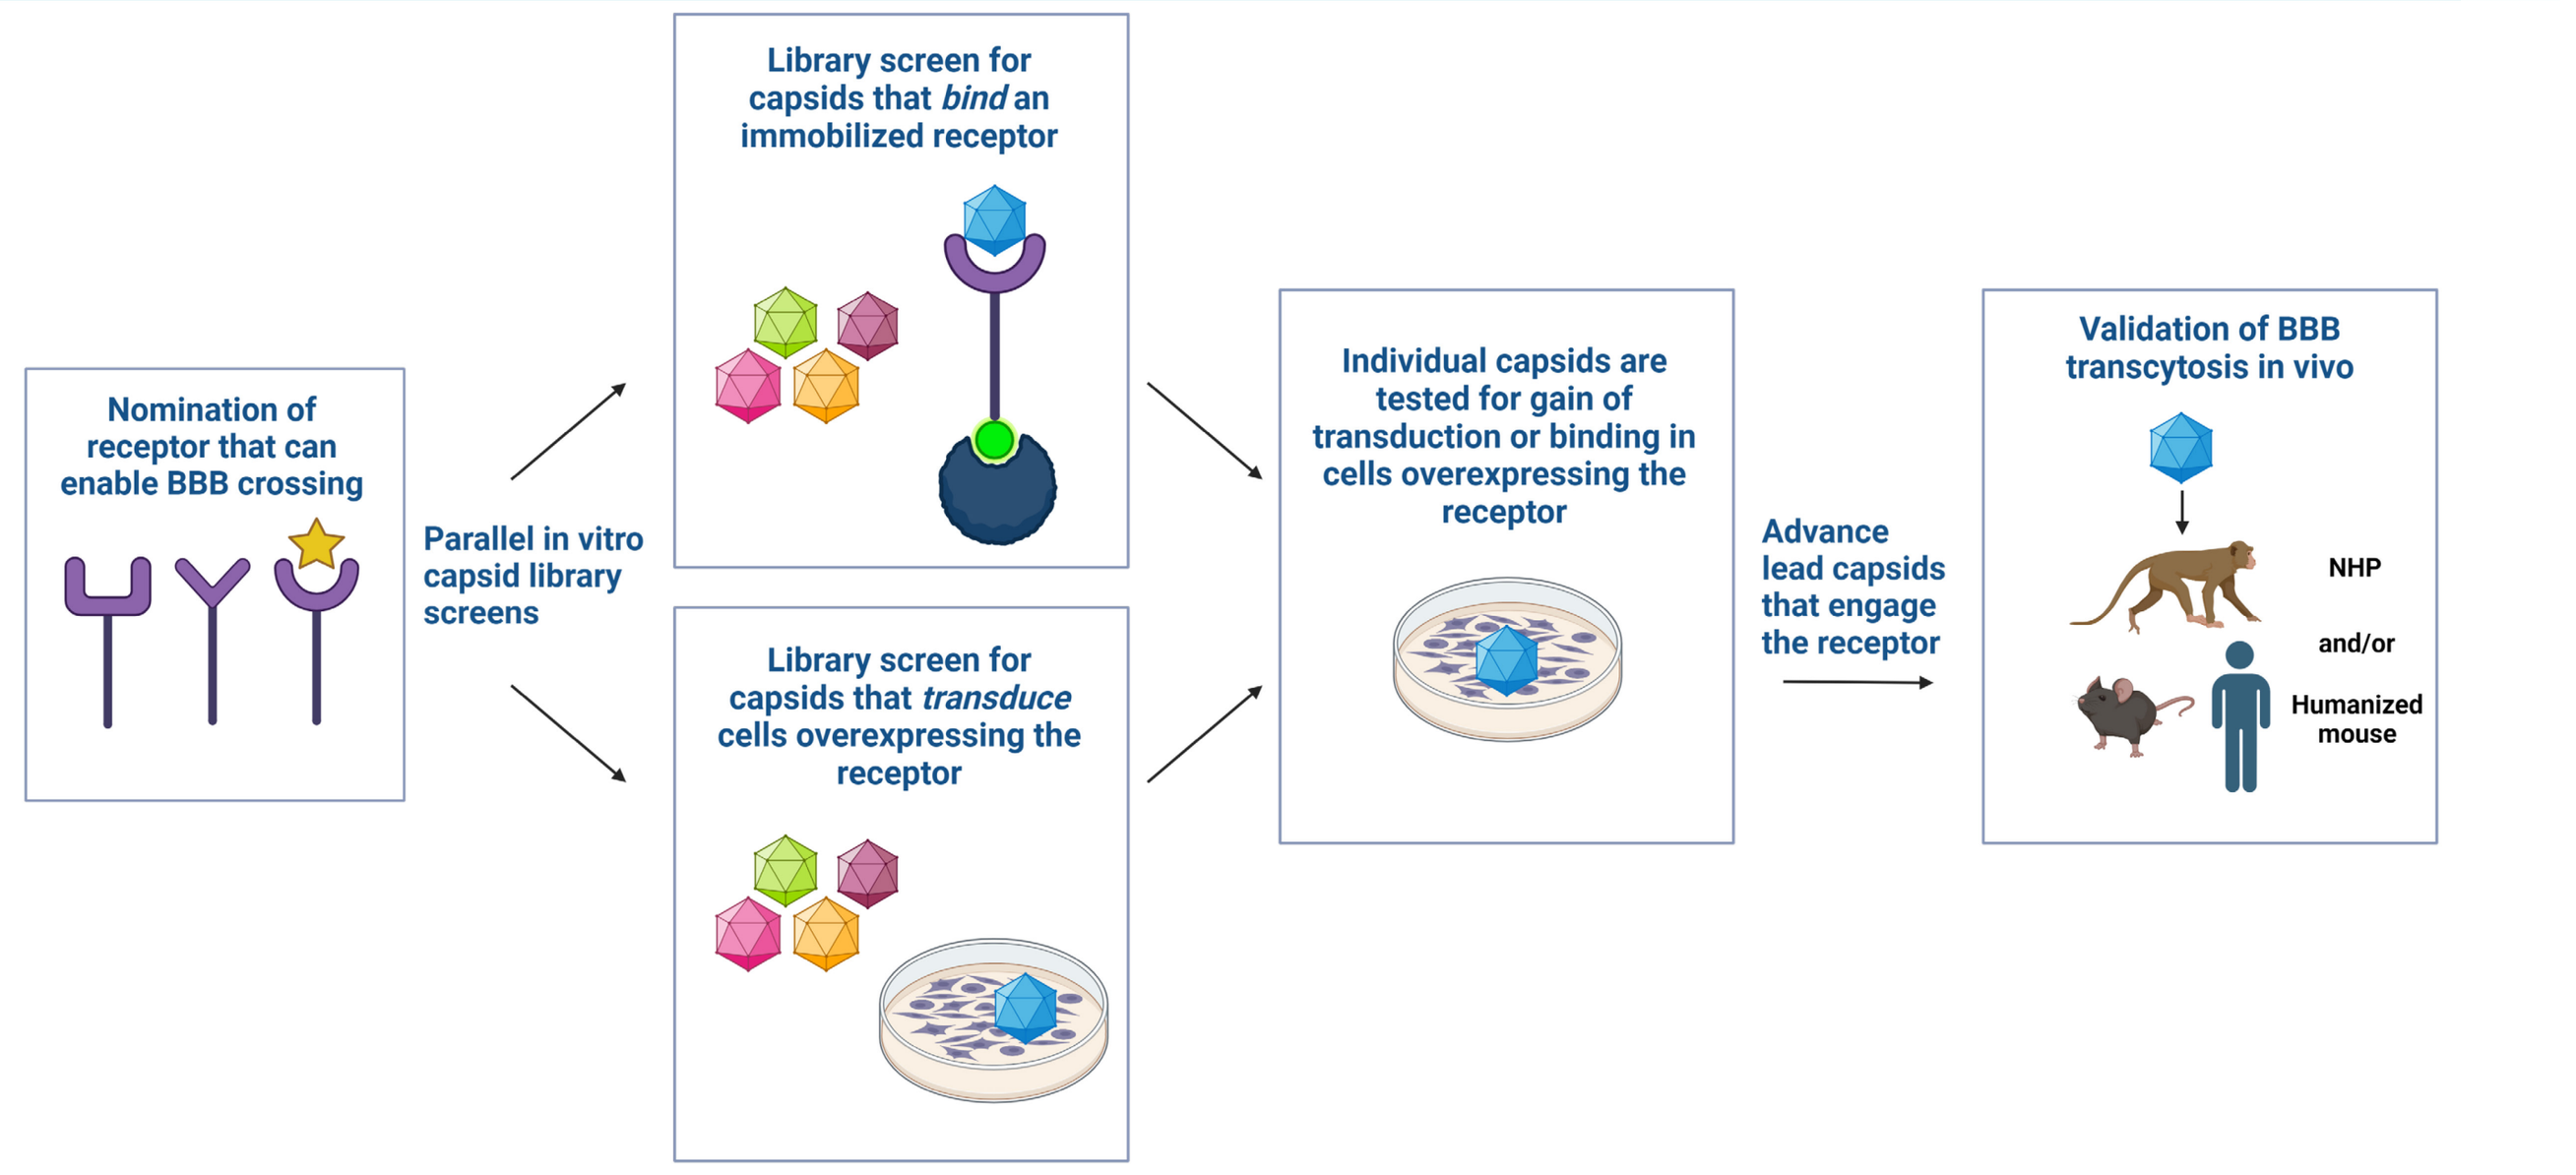
Nomination of receptor that can enable BBB crossing
Parallel in vitro capsid library screens
Library screen for capsids that *bind* an immobilized receptor
Library screen for capsids that *transduce* cells overexpressing the receptor
Individual capsids are tested for gain of transduction or binding in cells overexpressing the receptor
Advance lead capsids that engage the receptor
Validation of BBB transcytosis in vivo
NHP
and/or
Humanized mouse

Figure 1. Receptor-targeted capsid engineering approach. Capsid libraries are screened in parallel for binding to the immobilized receptor and for transduction of cells that overexpress the receptor. Capsids that specifically target the receptor are next evaluated individually to confirm targeting specificity and assess cross-species performance. Lead capsids are advanced for *in vivo* assessment of BBB crossing and CNS delivery in animal models.

Methods

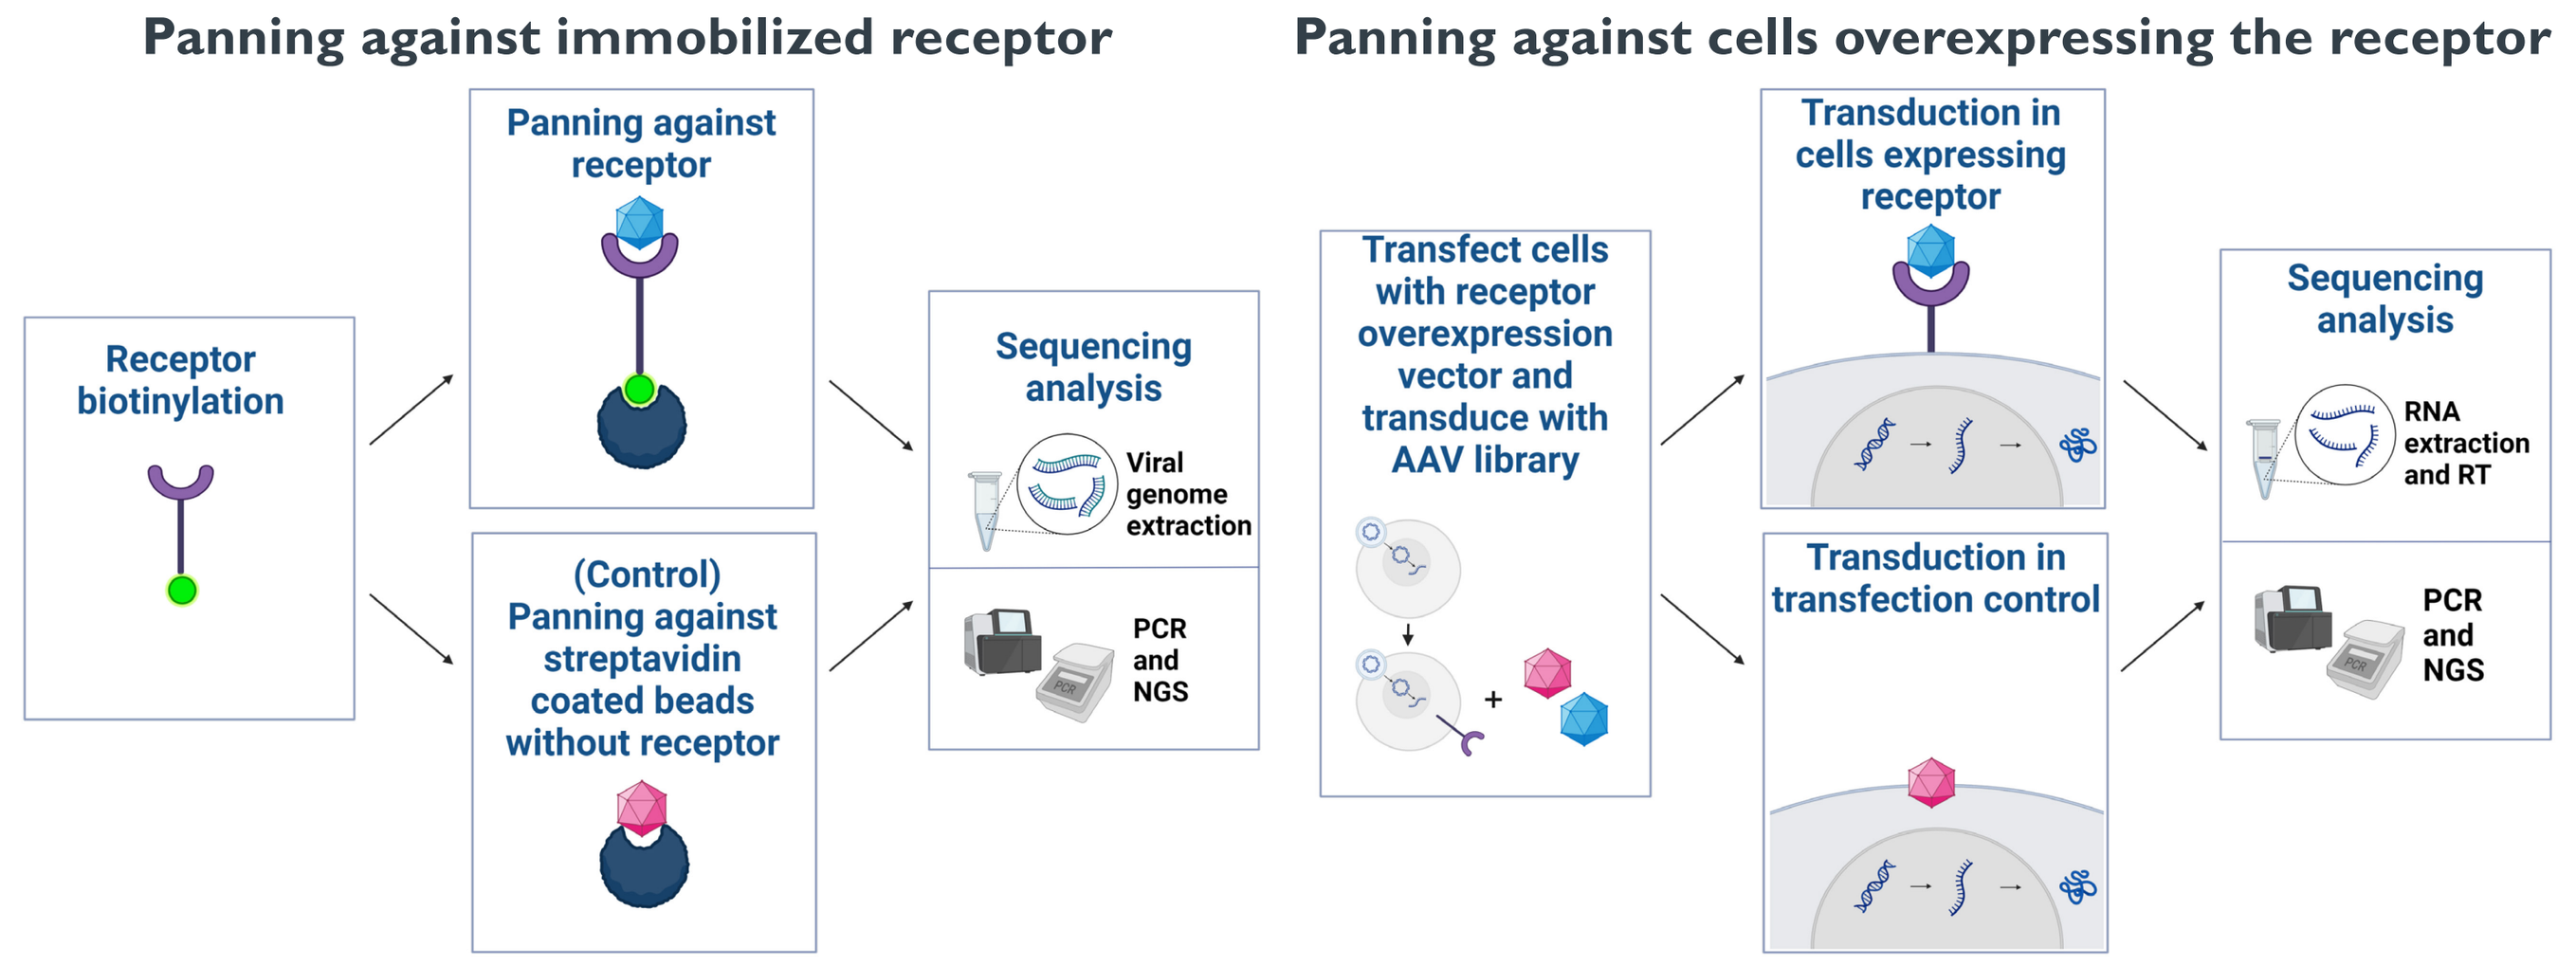
Panning against immobilized receptor
Panning against cells overexpressing the receptor
Receptor biotinylation
Panning against receptor
(Control) Panning against streptavidin coated beads without receptor
Sequencing analysis
Viral genome extraction
PCR and NGS
Transfect cells with receptor overexpression vector and transduce with AAV library
Transduction in cells expressing receptor
Transduction in transfection control
Sequencing analysis
RNA extraction and RT
PCR and NGS

Figure 2. Workflows for library screening. AAV capsid libraries are screened for receptor targeting relative to background binding in control samples. Capsid performance is assessed by next-generation sequencing and bioinformatic analysis is used to identify capsid variants that specifically target the receptor.

Library screens for capsids targeting Receptor 1

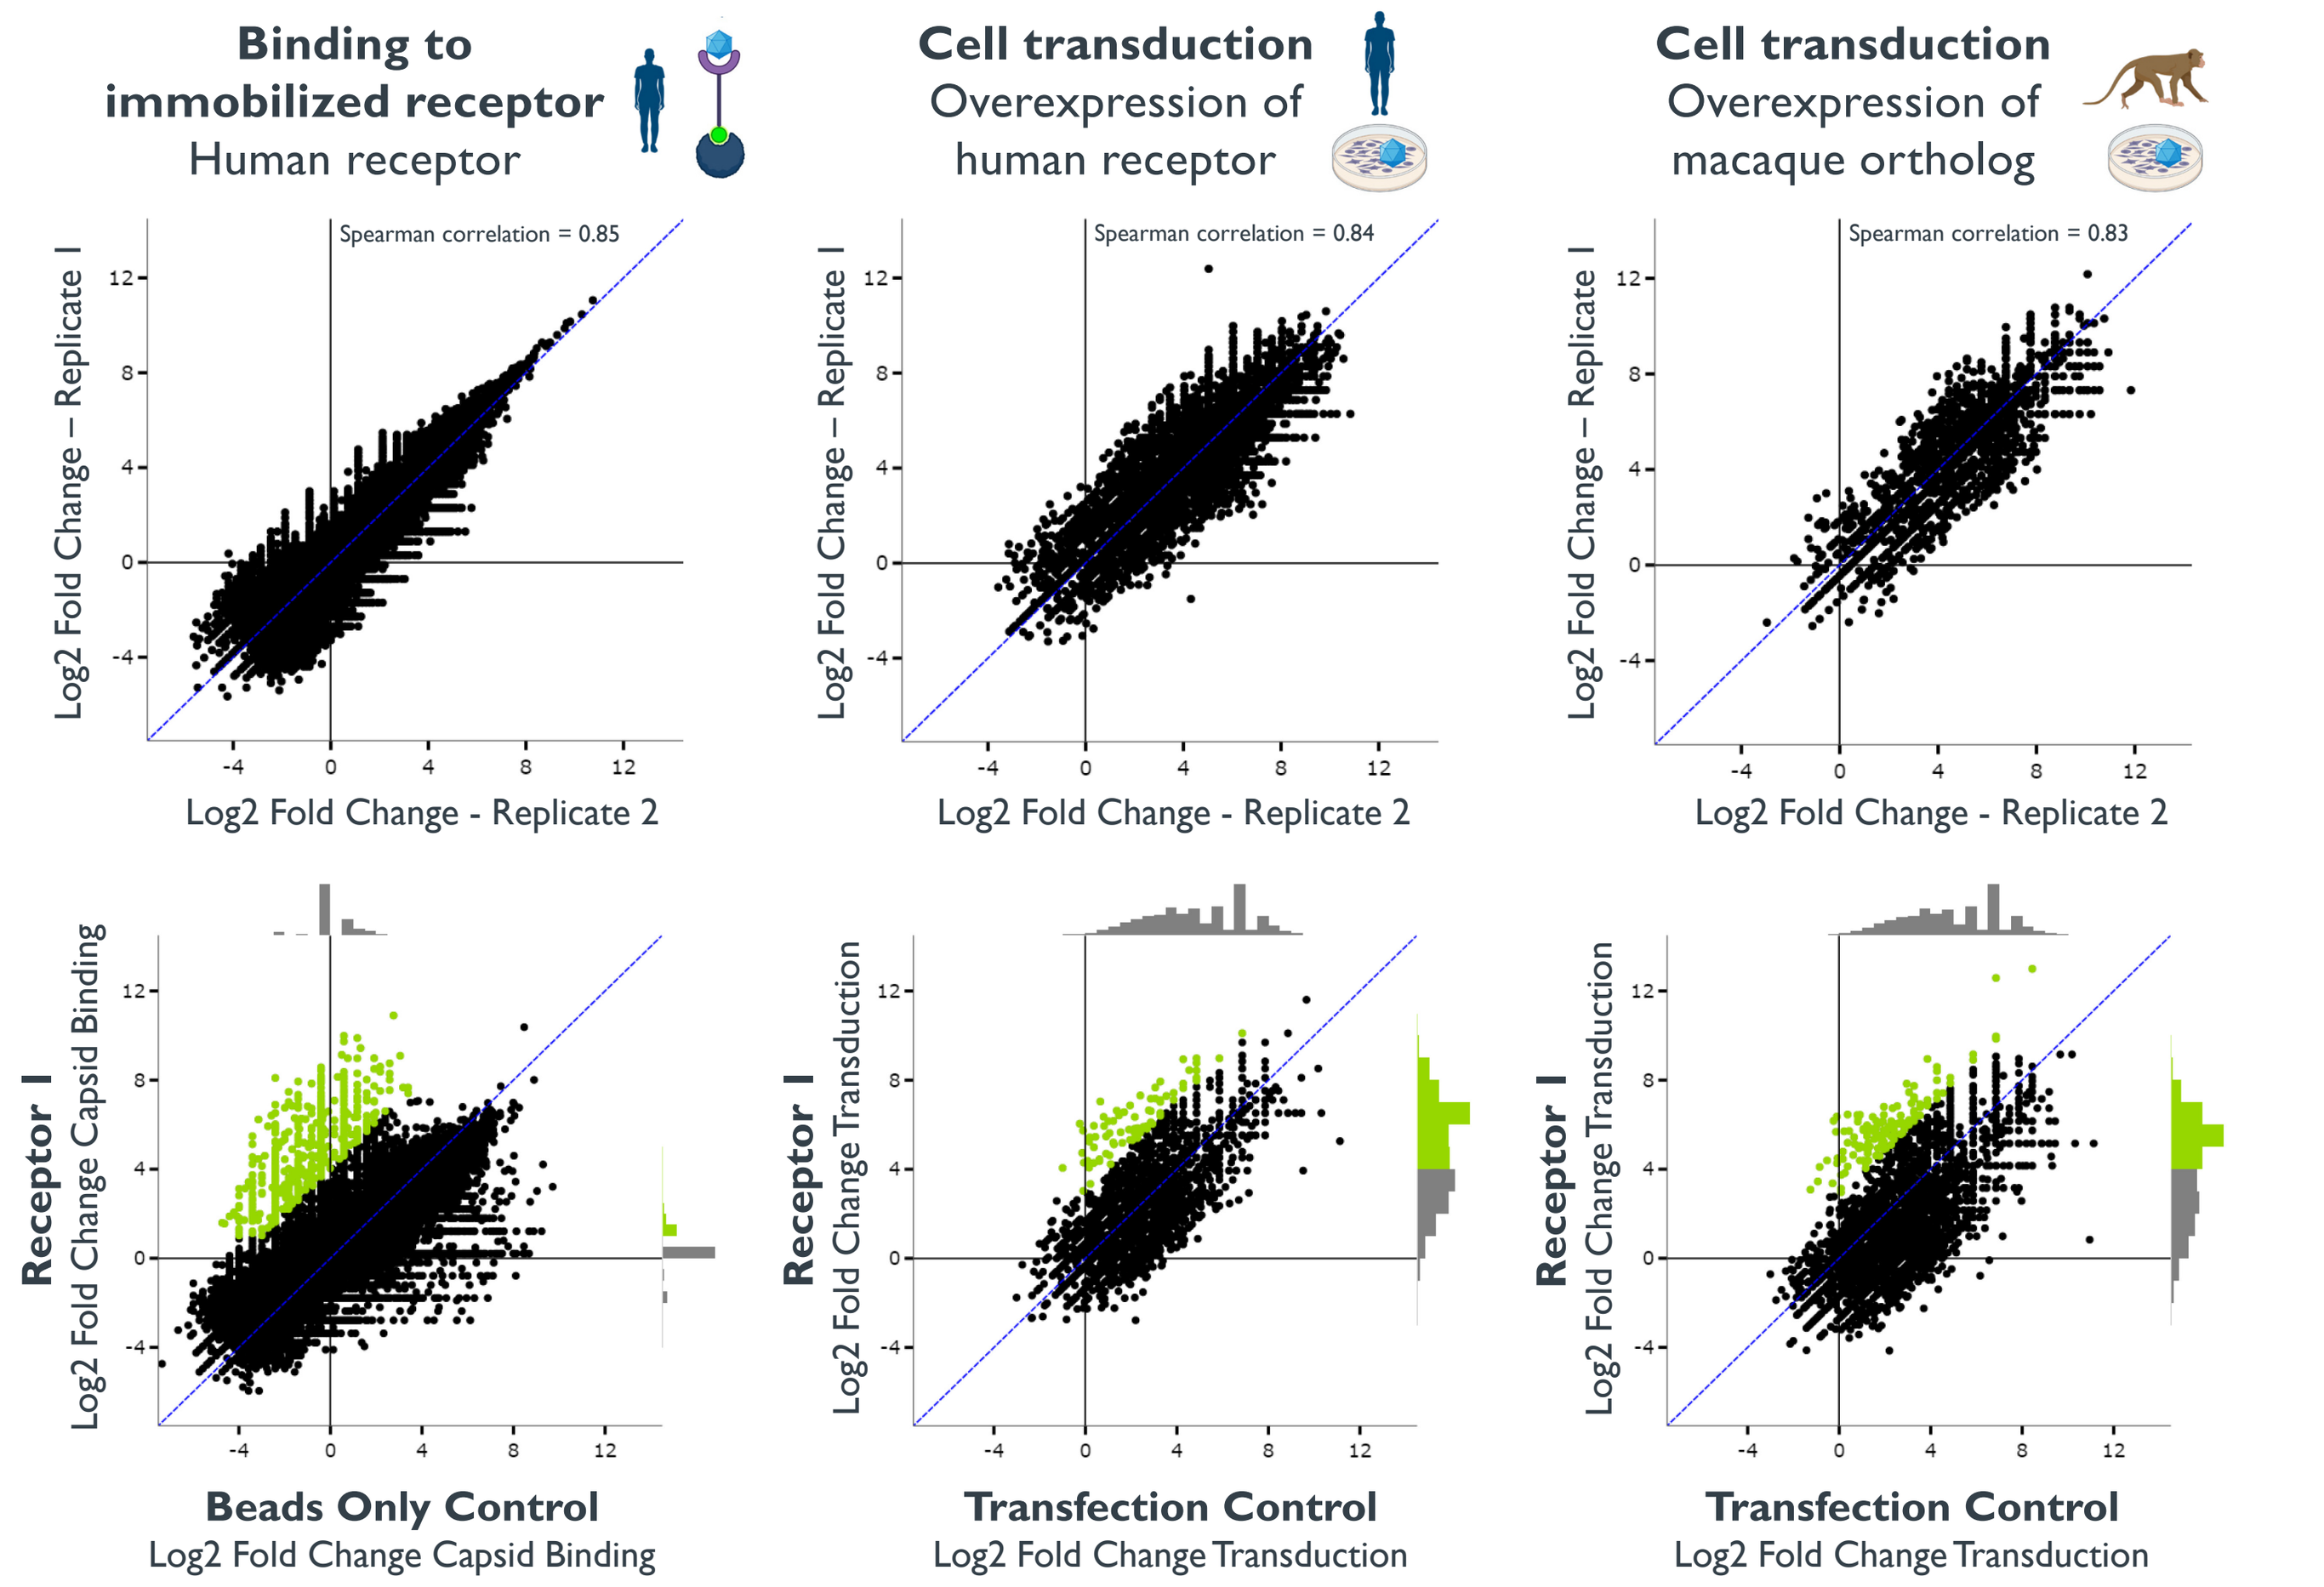
Binding to immobilized receptor
Human receptor
Cell transduction
Overexpression of human receptor
Cell transduction
Overexpression of macaque ortholog
Spearman correlation = 0.85
Spearman correlation = 0.84
Spearman correlation = 0.83
Log2 Fold Change – Replicate 1
Log2 Fold Change - Replicate 2
Receptor 1
Log2 Fold Change Capsid Binding
Log2 Fold Change Transduction
Beads Only Control
Transfection Control

Figure 3. Library screens identify capsid variants that specifically target Receptor 1. (Top) Biological replicates are highly correlated in library screens targeting Receptor 1. (Bottom) Three parallel capsid library screens were conducted:
1) Immobilized human receptor versus a bead only control.
2) Transduction of cells overexpressing the human receptor versus a transfection control.
3) Transduction of cells overexpressing the macaque receptor versus a transfection control.
Capsids exhibiting specific enrichment for Receptor 1 are colored in green.

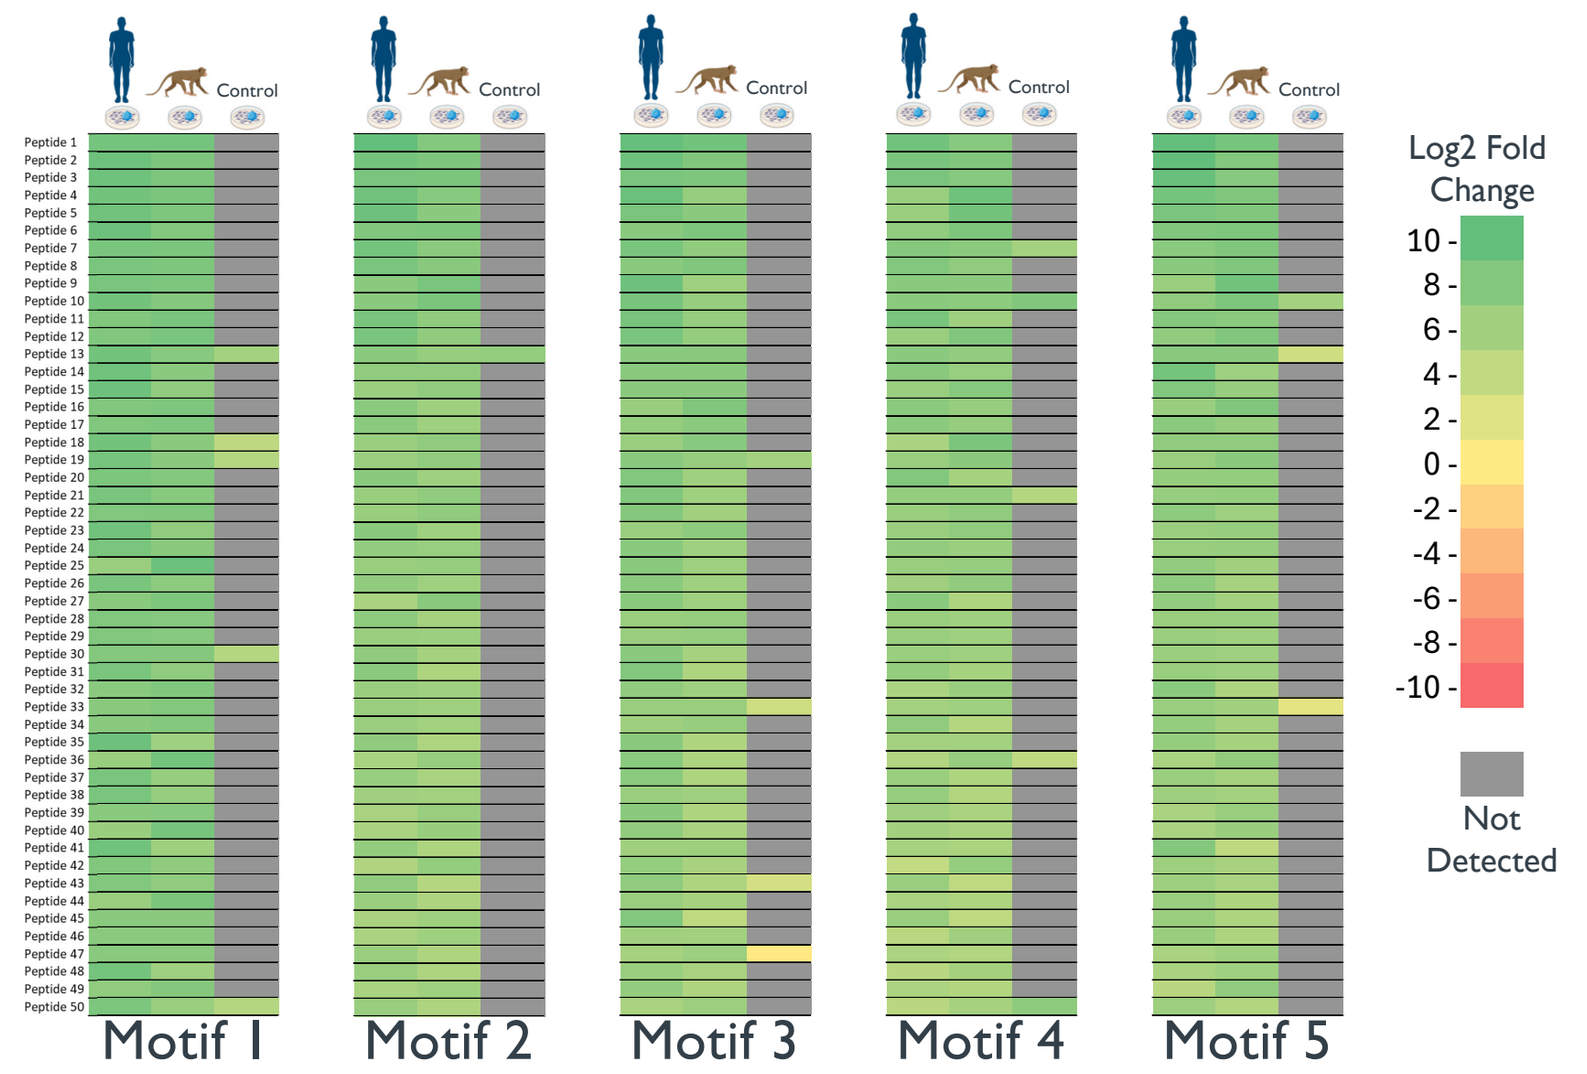
Control
Log2 Fold Change
Not Detected
Motif 1
Motif 2
Motif 3
Motif 4
Motif 5

Figure 4. Peptide motifs are enriched in engineered capsids targeting Receptor 1. The top 50 capsids with cross-species targeting of Receptor 1 are shown for each motif.

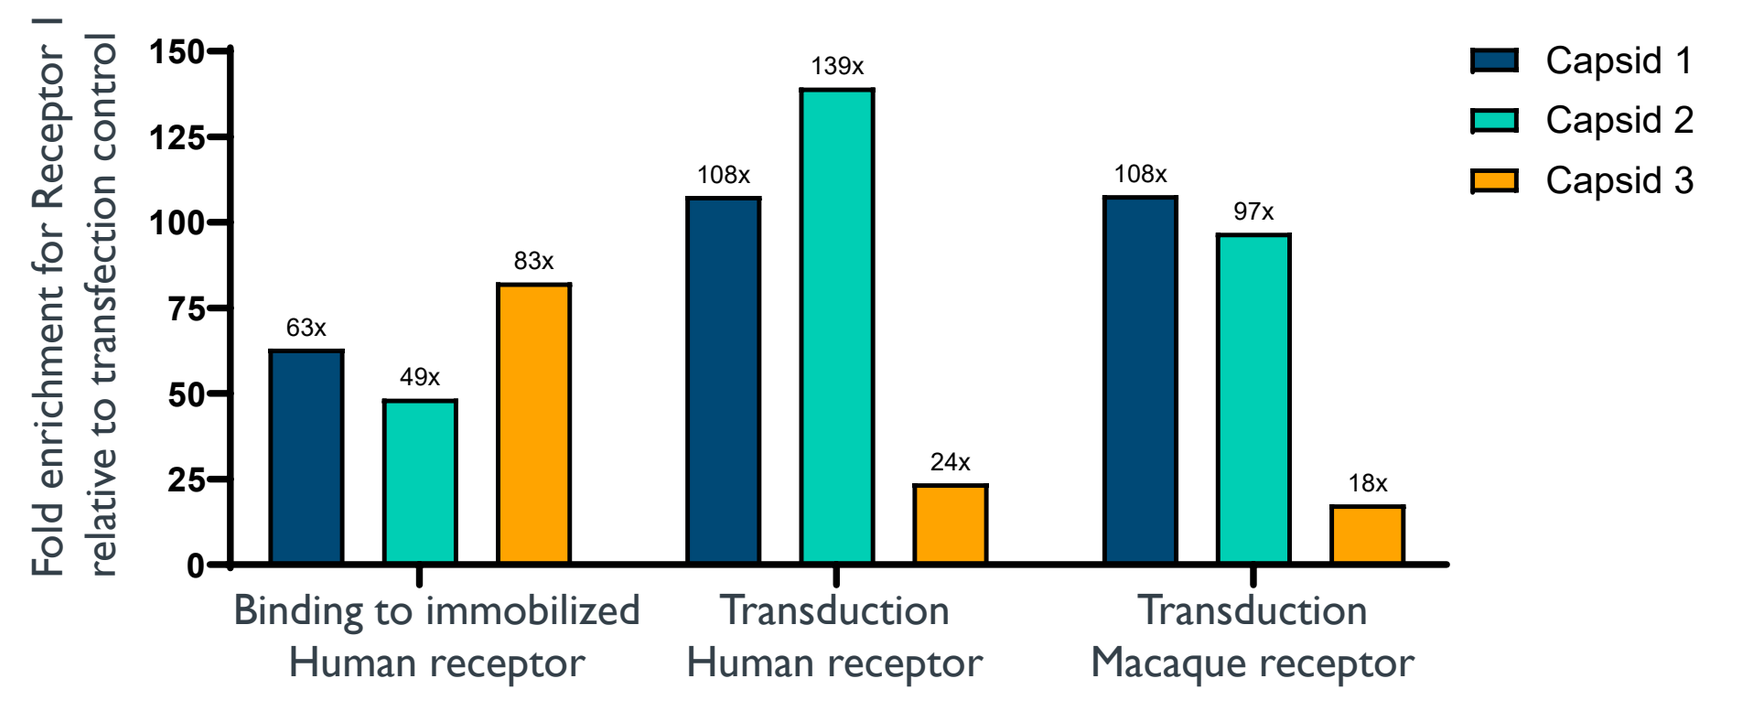
Fold enrichment for Receptor 1 relative to transfection control
Capsid 1
Capsid 2
Capsid 3
63x
49x
83x
108x
139x
24x
108x
97x
18x
Binding to immobilized Human receptor
Transduction Human receptor
Transduction Macaque receptor

Figure 5. Examples of capsids that exhibited enrichment in all three Receptor 1 screens and were advanced for subsequent individual evaluation experiments.

Individual evaluation of capsids targeting Receptor 1

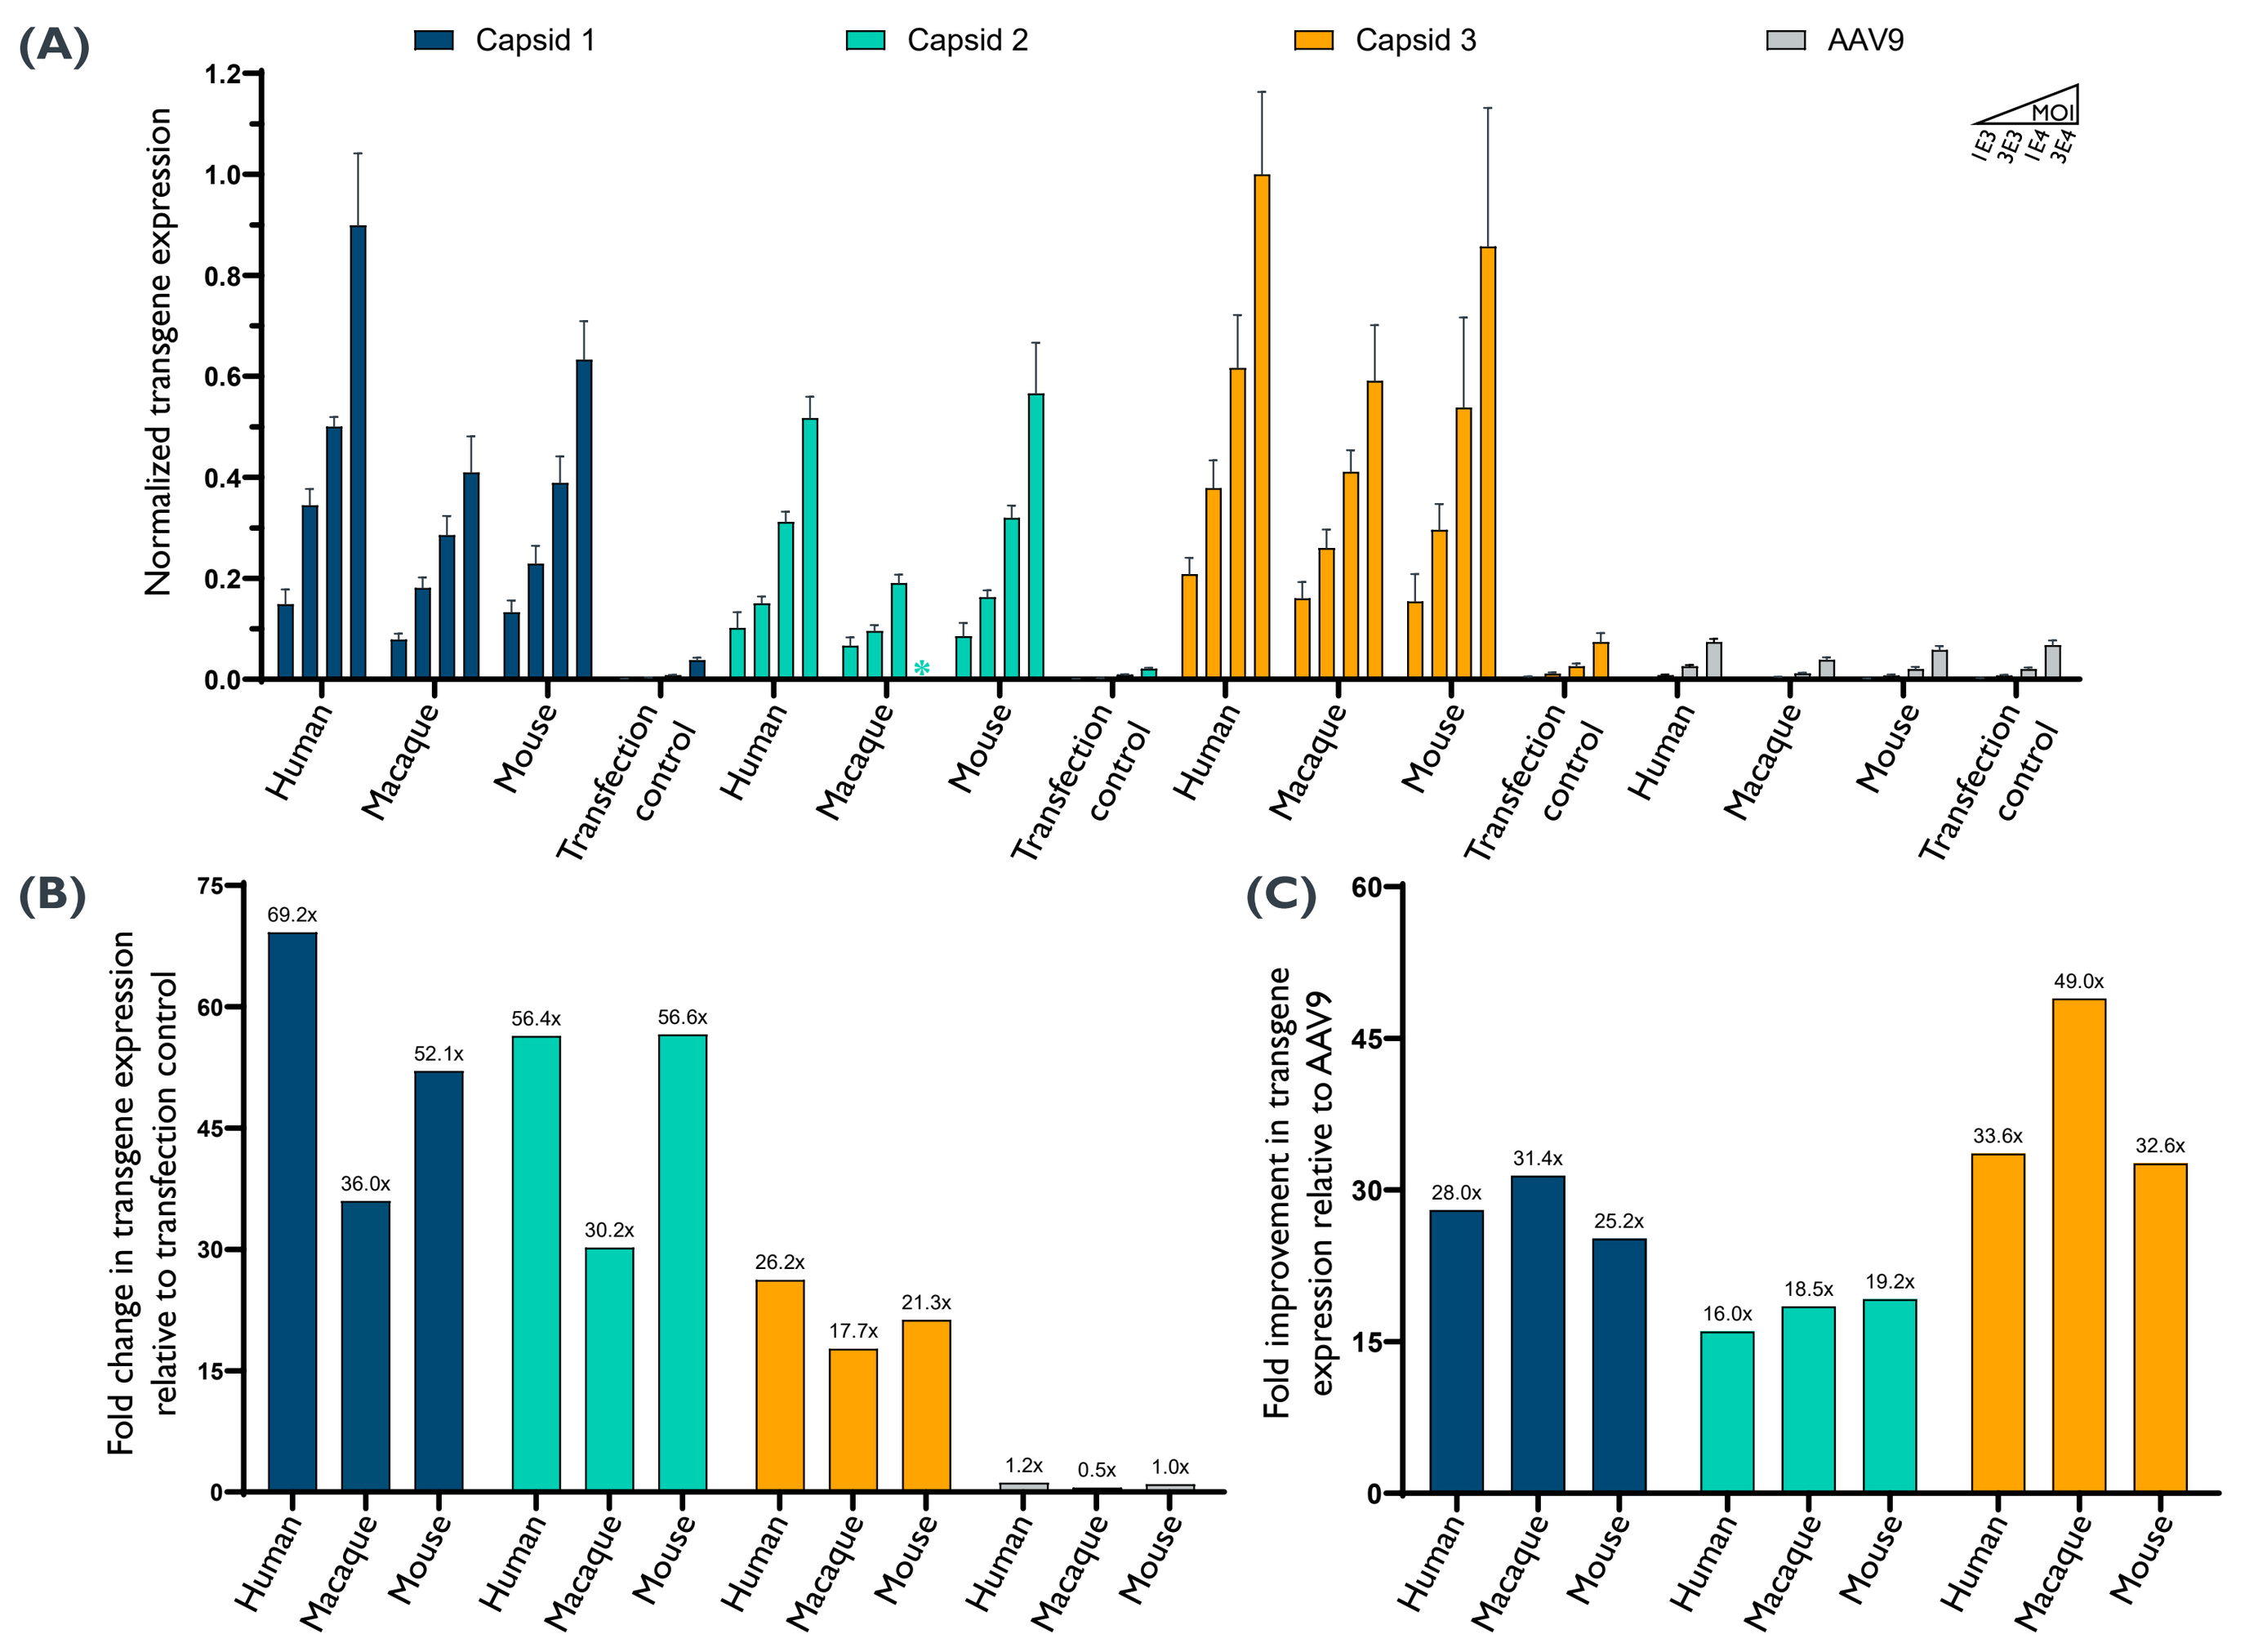
(A)
Capsid 1
Capsid 2
Capsid 3
AAV9
MOI
1E3
3E3
1E4
3E4
Normalized transgene expression
Human
Macaque
Mouse
Transfection control
(B)
Fold change in transgene expression relative to transfection control
69.2x
36.0x
52.1x
56.4x
30.2x
56.6x
26.2x
17.7x
21.3x
1.2x
0.5x
1.0x
(C)
Fold improvement in transgene expression relative to AAV9
28.0x
31.4x
25.2x
16.0x
18.5x
19.2x
33.6x
49.0x
32.6x

Figure 6. Engineered capsids exhibit enhanced transduction of cells overexpressing the human, macaque, or mouse ortholog of Receptor 1.
Cells were transiently transfected with an overexpression plasmid encoding Receptor 1. Transfected cells were transduced with each AAV and transgene mRNA expression was assessed 72 hours later by RT-qPCR. *Sample lost during processing.

(A) Capsids 1-3 were evaluated individually and compared against the parental serotype AAV9. Receptor-targeted capsids exhibit a marked enhancement in transgene mRNA expression in cells overexpressing Receptor 1, but not in the transfection control condition. Moreover, this gain of function was conserved across human, macaque, and mouse orthologs of Receptor 1.

(B) Fold increase in transgene expression relative to control. A nonlinear regression model was used to interpolate relative transgene expression values for each capsid-receptor condition. These values were then scaled to the transfection control value for each capsid.

(C) Engineered capsids mediate higher transgene expression than AAV9 in cells overexpressing human, macaque, or mouse Receptor 1.

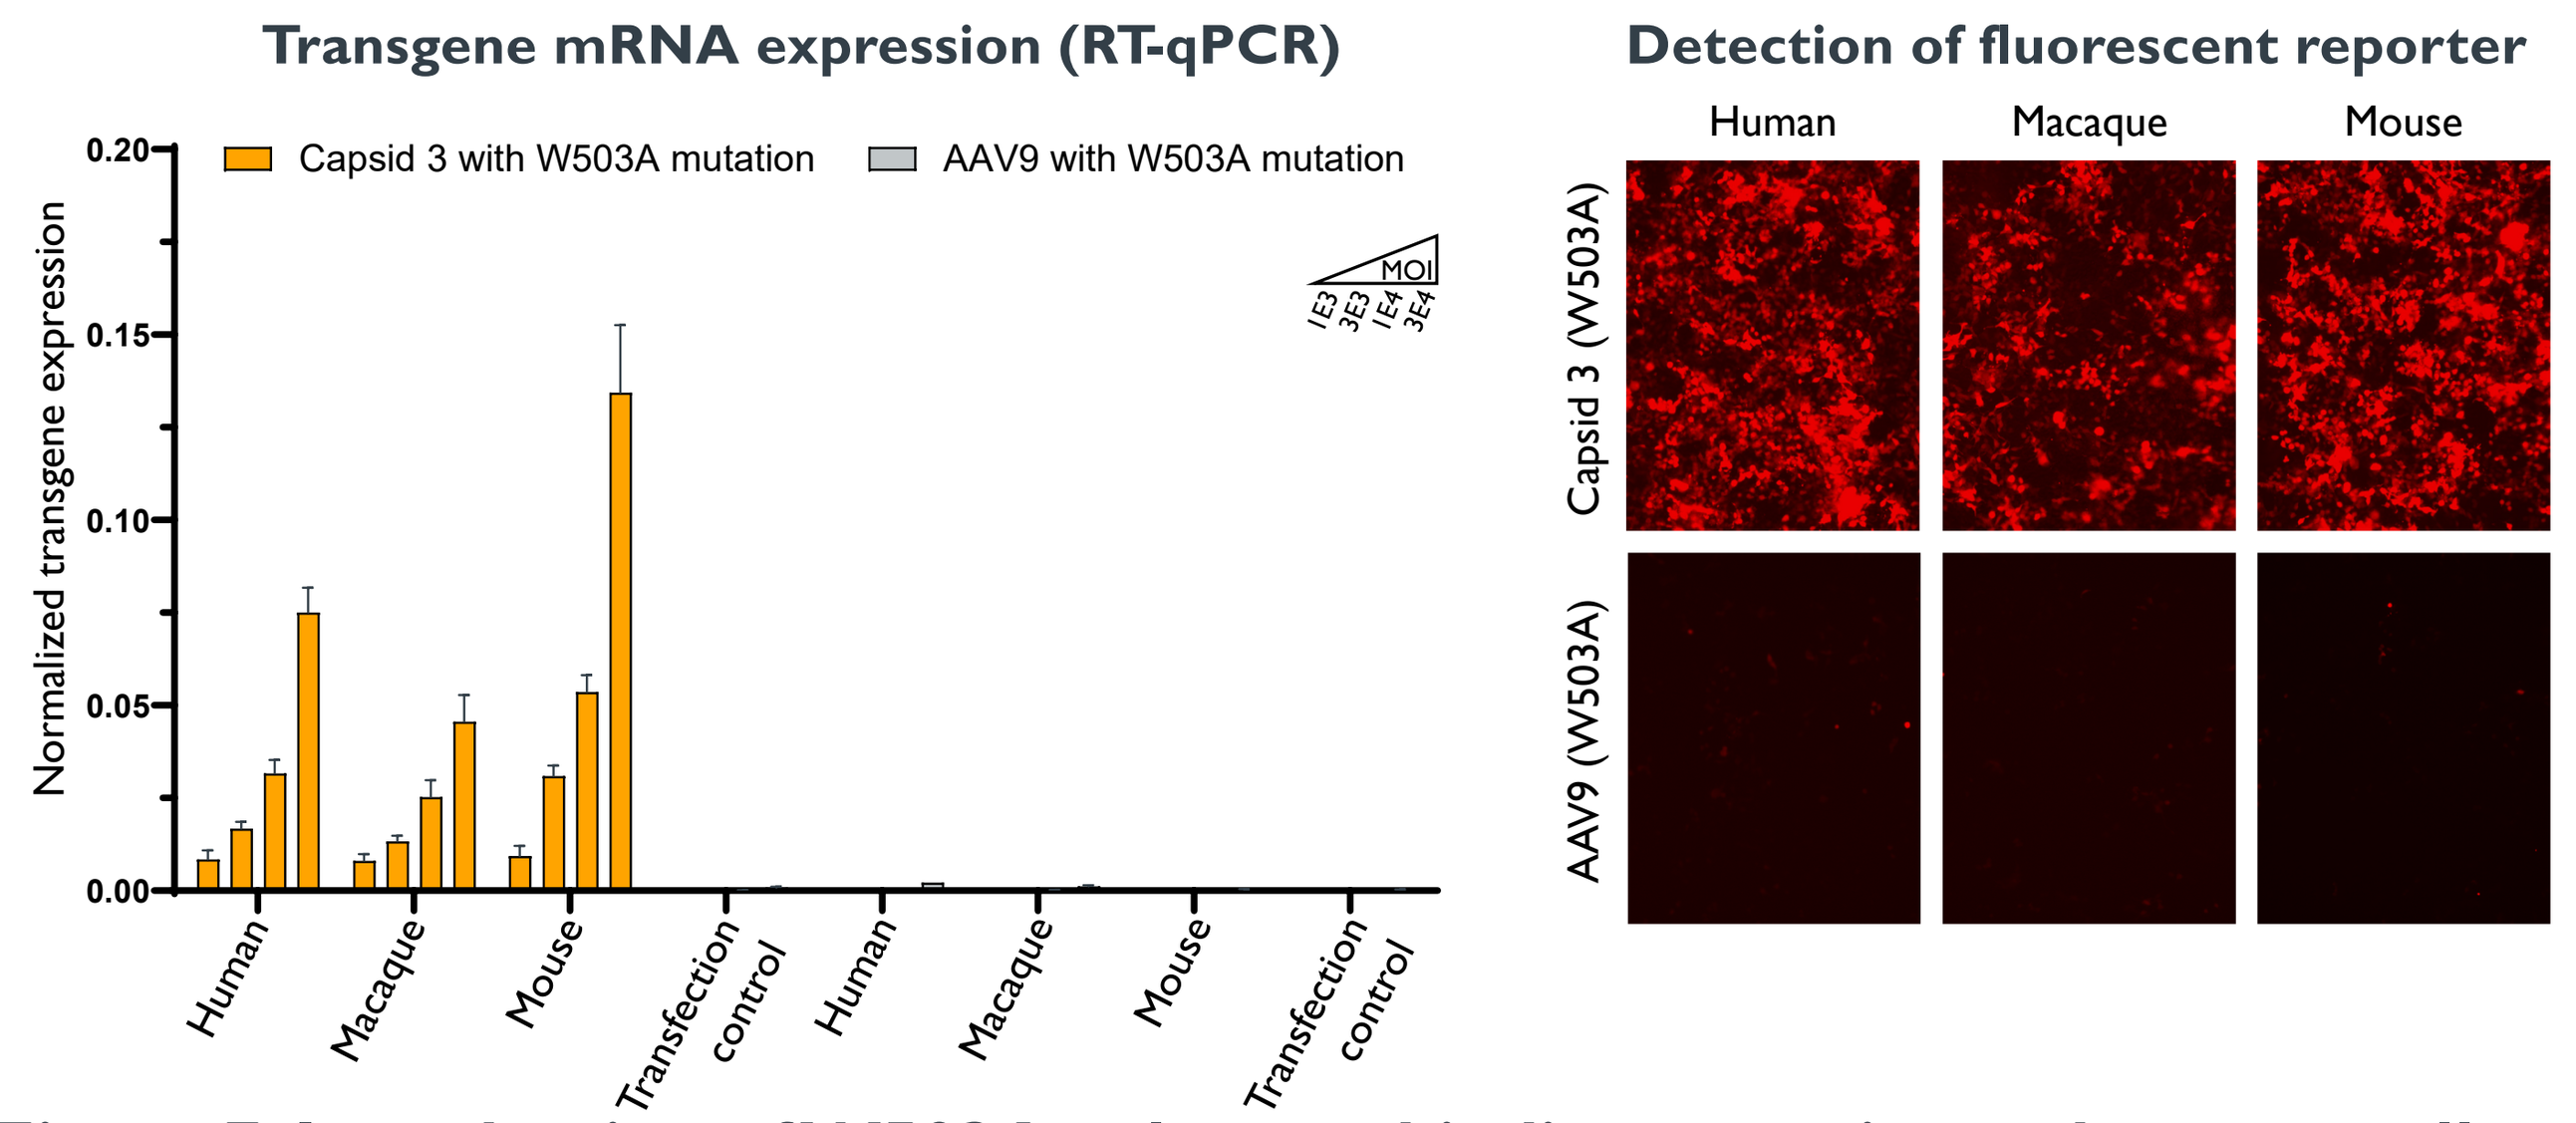
Transgene mRNA expression (RT-qPCR)
Capsid 3 with W503A mutation
AAV9 with W503A mutation
MOI
1E3
3E3
1E4
3E4
Normalized transgene expression
Human
Macaque
Mouse
Transfection control
Detection of fluorescent reporter
Human
Macaque
Mouse
Capsid 3 (W503A)
AAV9 (W503A)

Figure 7. Introduction of W503A galactose binding mutation reduces overall potency, but a gain of function remains in cells overexpressing Receptor 1. These results suggest that Capsid 3 can utilize both galactose and Receptor 1 for cell entry.

Library screens for capsids targeting Receptor 2 or 3

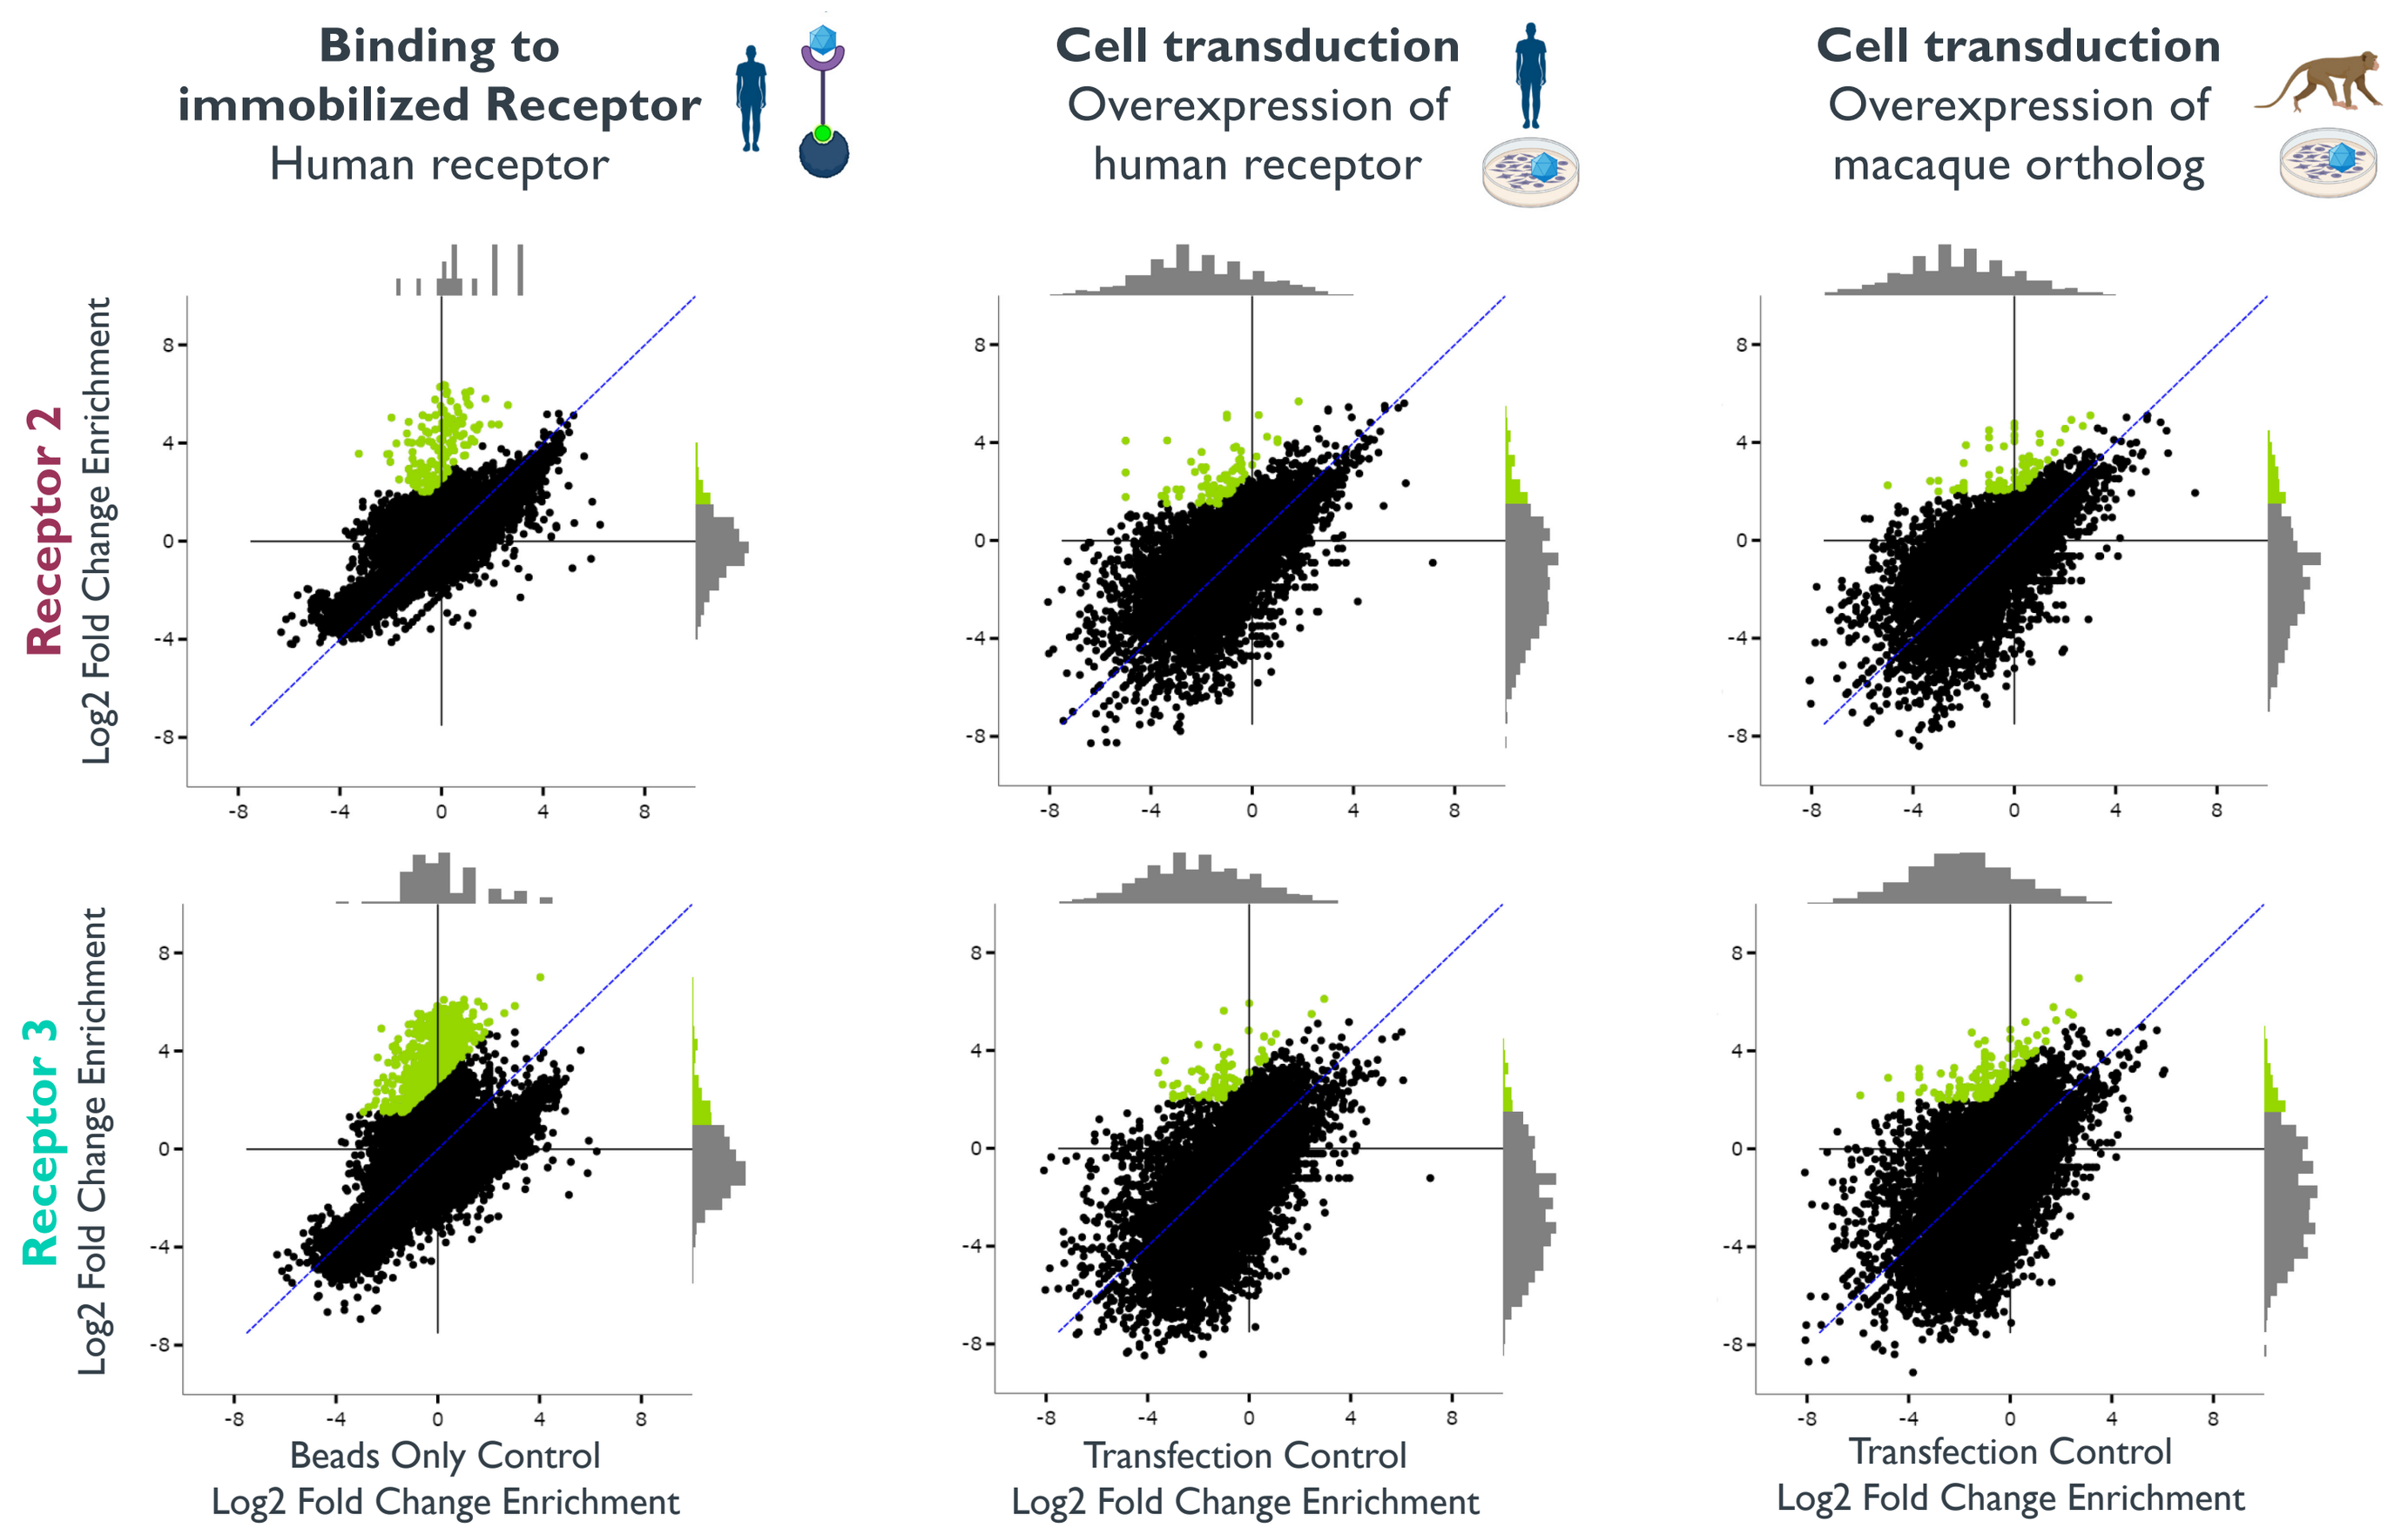
Binding to immobilized Receptor
Human receptor
Cell transduction
Overexpression of human receptor
Cell transduction
Overexpression of macaque ortholog
Receptor 2
Receptor 3
Log2 Fold Change Enrichment
Beads Only Control
Transfection Control

Figure 8. Library screens identify capsid variants that specifically target Receptor 2 or Receptor 3. Three parallel capsid library screens were conducted for each receptor:
1) Immobilized human receptor versus a bead only control.
2) Transduction of cells overexpressing the human receptor versus a transfection control.
3) Transduction of cells overexpressing the macaque receptor versus a transfection control.
Capsids exhibiting specific enrichment for Receptor 2 or 3 are colored in green.

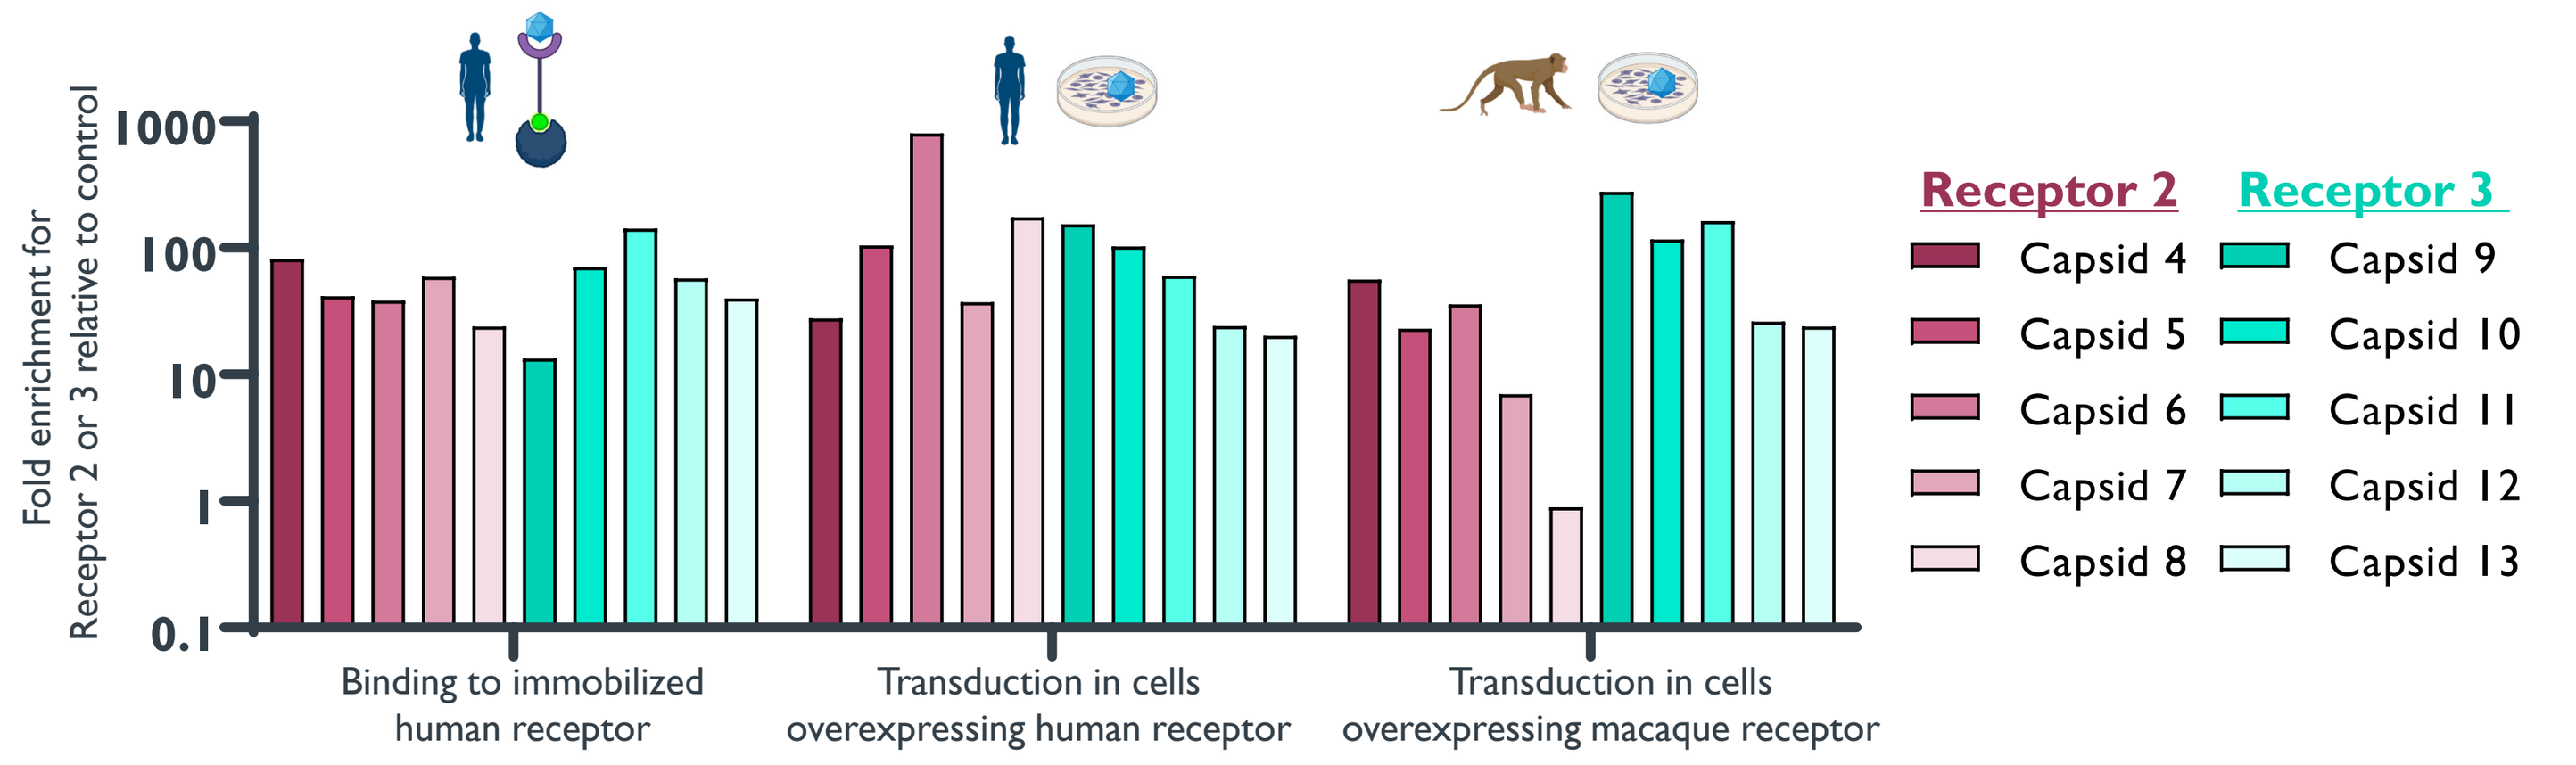
Fold enrichment for Receptor 2 or 3 relative to control
Receptor 2
Receptor 3
Capsid 4
Capsid 5
Capsid 6
Capsid 7
Capsid 8
Capsid 9
Capsid 10
Capsid 11
Capsid 12
Capsid 13
Binding to immobilized human receptor
Transduction in cells overexpressing human receptor
Transduction in cells overexpressing macaque receptor

Figure 9. Examples of capsids that exhibited enrichment in Receptor 2 or Receptor 3 screens and are being advanced for individual evaluation. A subset of identified capsids exhibit cross-species interaction with both the human and macaque orthologs.

Conclusions and next steps

- We have engineered receptor-targeted AAV capsids for three receptors that have the potential to mediate transport across the blood-brain barrier.
- Capsids targeting Receptor 1 exhibit enhanced transduction in cells overexpressing the human, macaque, and mouse orthologs. A subset of capsids targeting Receptors 2 and 3 display cross-species interaction with both the human and macaque orthologs.
- Receptor-targeted capsids will be evaluated *in vivo* to assess BBB crossing efficiency and tropism for CNS cell types.
- These receptor-targeted capsid families are distinct from STAC-BBB and likewise have the potential to mediate widespread CNS delivery for the treatment of neurological disorders.

Select figures created with BioRender.com

Presented at ASGCT 2024