3461

Updated Results of the Alta Study, a Phase 1/2 Study of Giroctocogene Fitelparvovec (PF-07055480/SB-525) Gene Therapy in Adults With Severe Hemophilia A

Adam Giermasz[1]*, Nathan Visweshwar[2], Thomas J Harrington[3], Andrew D Leavitt[4], Barbara A Konkle[5], Jeremy Rupon[6], Gregory Di Russo[7], Li-Jung Tseng[8], Maria de los Angeles Resa[8], Florence Ganne[9], Delphine Agathon[9], Frank Plonski[6], Didier Rouy[10*], Bettina M Cockroft[10], Annie Fang[8], Steven Arkin[7]; *Presenting author

[1]University of California Davis, Sacramento, CA, USA; [2]University of South Florida, Tampa, FL, USA; [3]University of Miami Miller School of Medicine, Miami, FL, USA; [4]University of California San Francisco, San Francisco, CA, USA; [5]University of Washington, Seattle, WA, USA; [6]Pfizer Inc, Collegeville, PA, USA; [7]Pfizer Inc, Cambridge, MA, USA; [8]Pfizer Inc, New York, NY, USA; [9]Pfizer Inc, Paris, France; [10]Sangamo Therapeutics, Brisbane, CA, USA; *At the time of the study; currently at Sanofi SA, Waltham, MA, USA

BACKGROUND

- Hemophilia A is an X-linked (*F8* gene) disorder of hemostasis that results in insufficient endogenous factor VIII (FVIII) activity.
- Current treatment involves replacement therapy with exogenous FVIII or treatment with a humanized bispecific antibody that simultaneously binds factor IX and X, both of which require frequent dosing via intravenous (IV) or subcutaneous (SC) administration.[1]
- Maintenance of FVIII activity in the mild (>5% to >40%) to normal (>50%) range improves outcomes for patients with severe hemophilia A (FVIII activity <1%).[2]
- This wide therapeutic window and the underlying defect of the *F8* gene make hemophilia A an ideal candidate for gene therapy.[3]
- Adeno-associated virus (AAV)–mediated gene transfer enables the delivery of a modified functional *F8* coding sequence to hepatocytes, allowing for the synthesis of endogenous FVIII at levels preventing unprovoked bleeding events.
- Giroctocogene fitelparvovec (PF-07055480, previously called SB-525) is a liver-tropic recombinant AAV serotype 6 (rAAV6) vector encoding a modified B-domain–deleted *F8* coding sequence that is in development for the treatment of hemophilia A (**Figure 1**).

Figure 1. Giroctocogene Fitelparvovec Gene Therapy

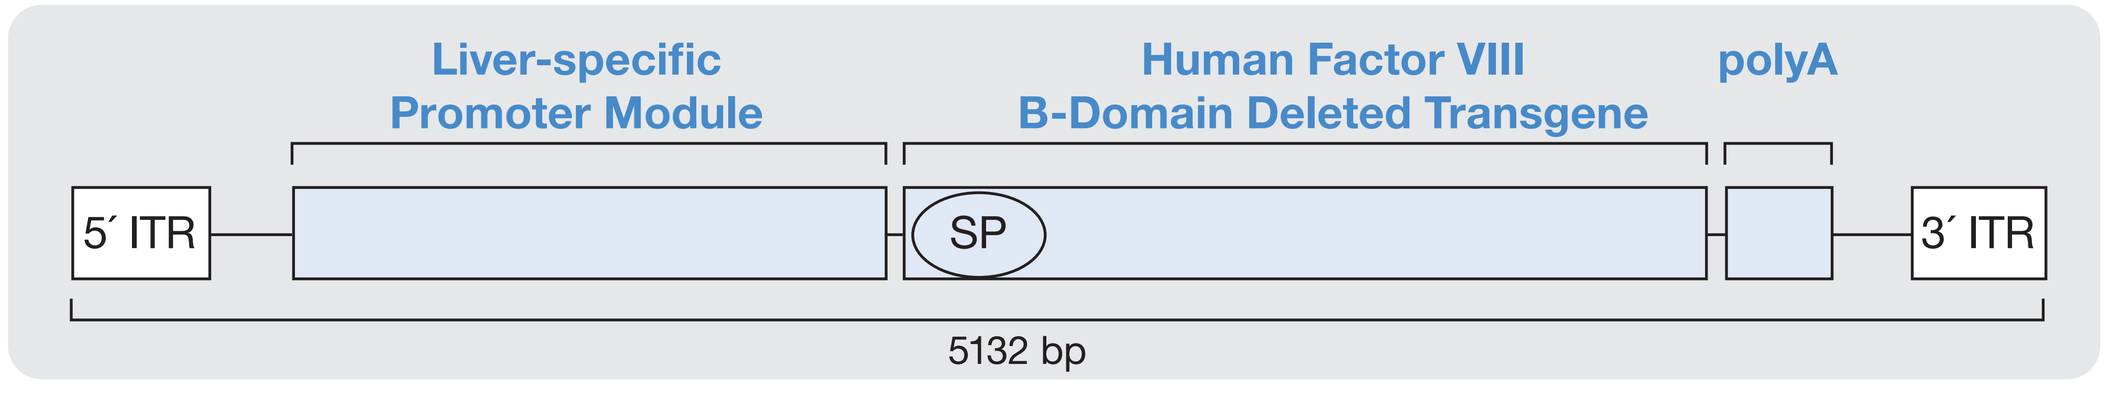
Liver-specific
Promoter Module
Human Factor VIII
B-Domain Deleted Transgene
polyA
5´ ITR
SP
3´ ITR
5132 bp

bp=base pairs; ITR=internal tandem repeat; SP=signal peptide

OBJECTIVE

- To present updated results with 3 years of follow-up on all participants from Alta, an ongoing gene therapy study in patients with severe hemophilia A treated with giroctocogene fitelparvovec.

METHODS

Study Design

- Alta (NCT03061201) is a phase 1/2, multicenter, dose-ranging study to assess the safety and tolerability of giroctocogene fitelparvovec (**Figure 2**).

Figure 2. Alta Participants and Study Design

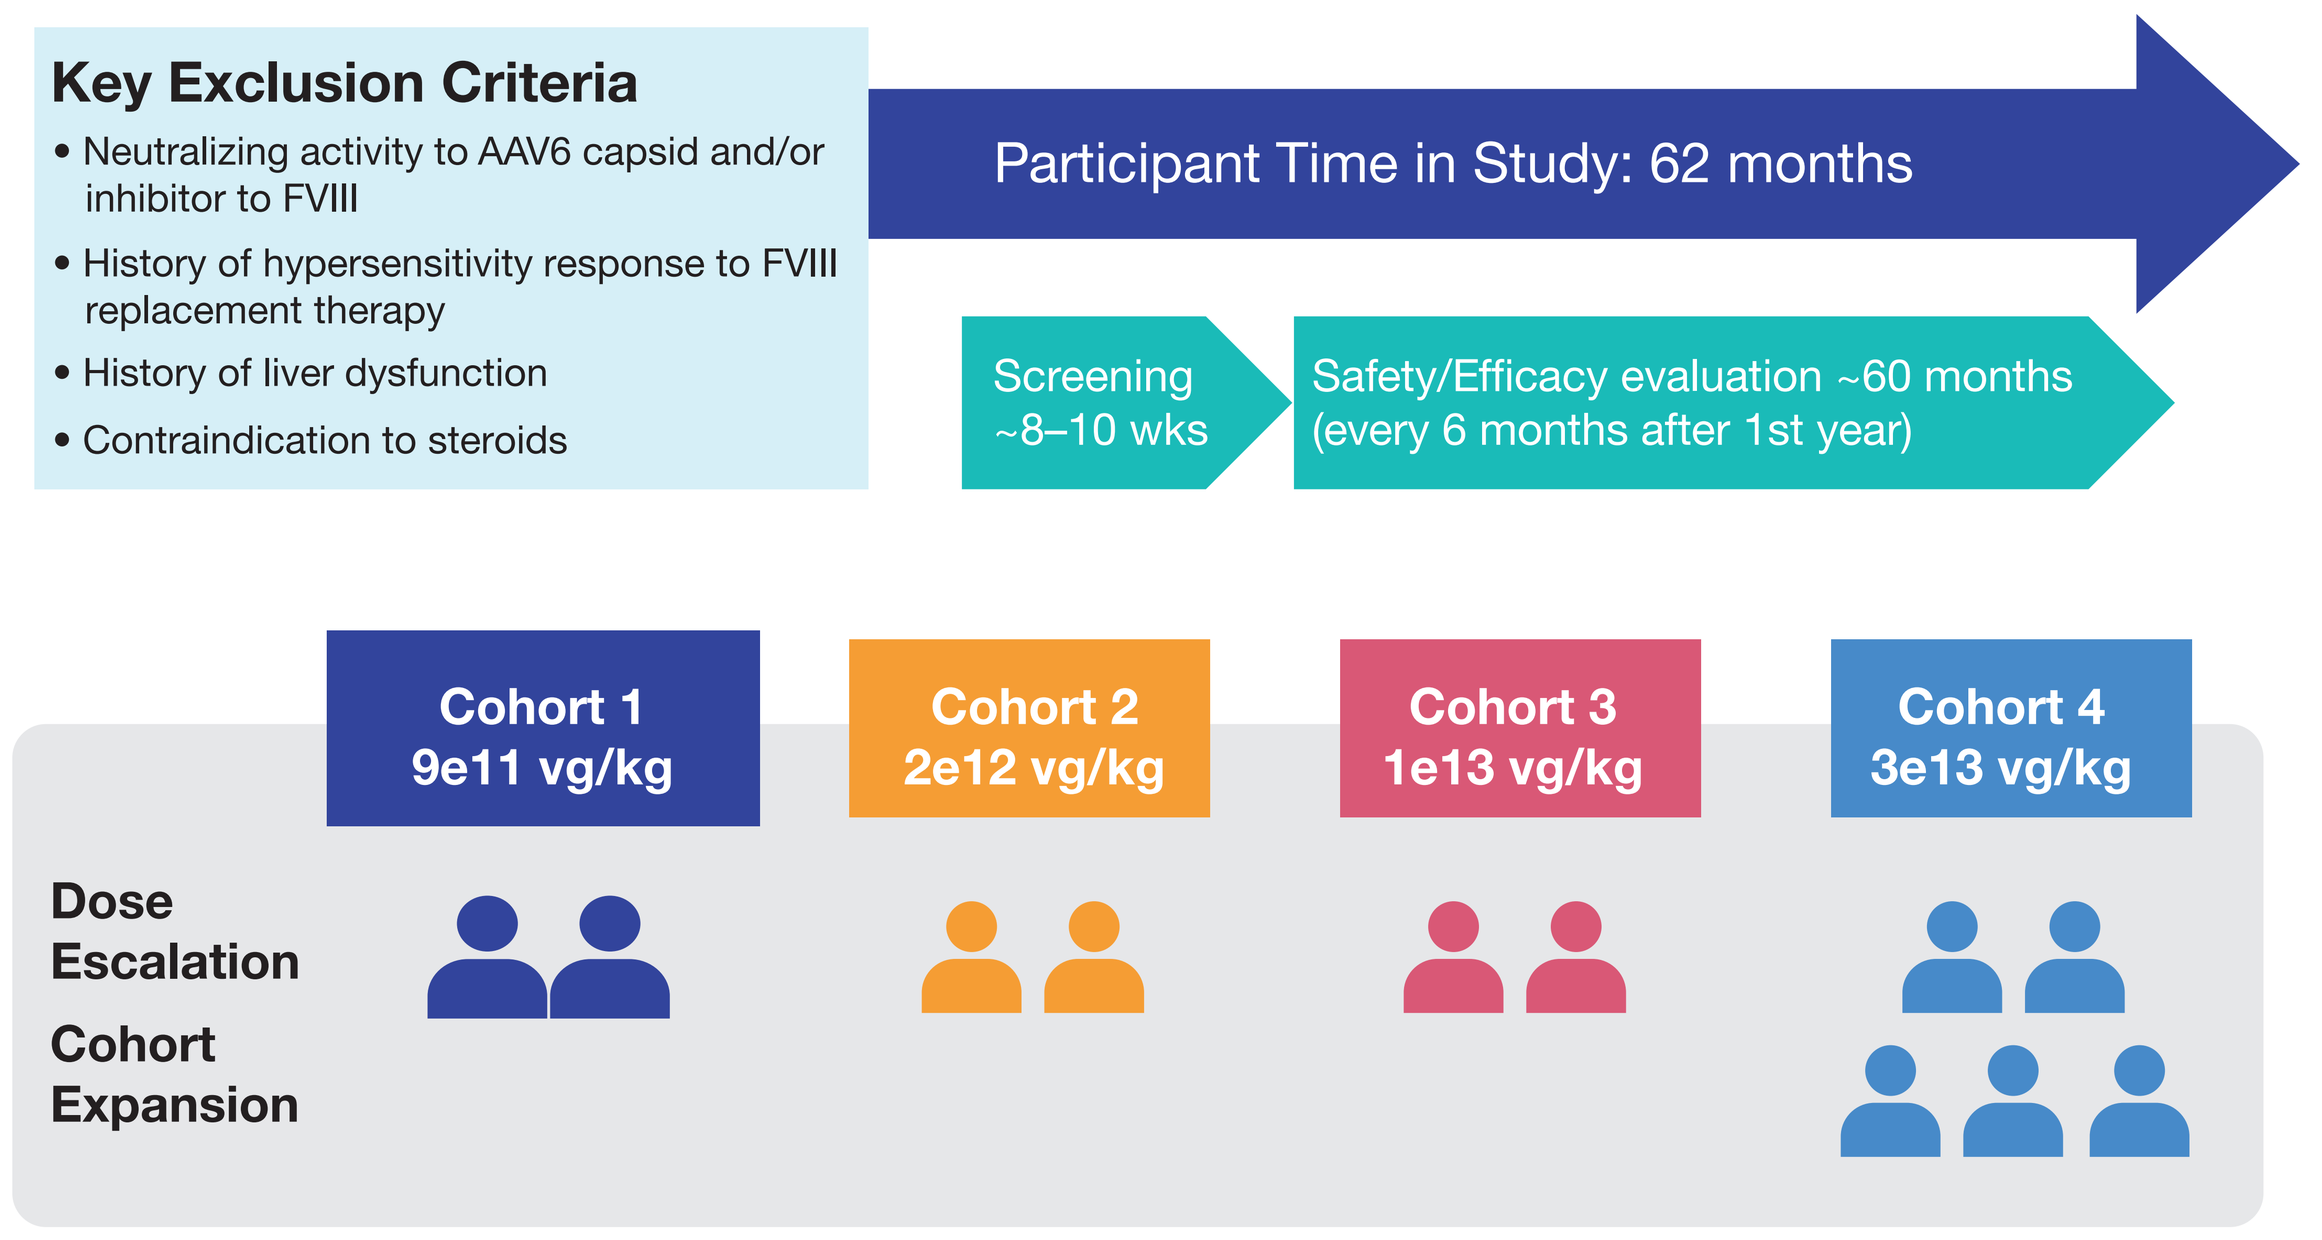
Key Exclusion Criteria
• Neutralizing activity to AAV6 capsid and/or inhibitor to FVIII
• History of hypersensitivity response to FVIII replacement therapy
• History of liver dysfunction
• Contraindication to steroids
Participant Time in Study: 62 months
Screening ~8–10 wks
Safety/Efficacy evaluation ~60 months (every 6 months after 1st year)
Cohort 1 9e11 vg/kg
Cohort 2 2e12 vg/kg
Cohort 3 1e13 vg/kg
Cohort 4 3e13 vg/kg
Dose Escalation
Cohort Expansion

AAV6=adeno-associated virus serotype 6; FVIII=coagulation factor VIII; vg=vector genomes

Participant Population

- Adults (aged ≥18 years) with severe hemophilia A (FVIII activity <1%).
- Key Exclusion criteria are shown in **Figure 2**.

Trial Procedures

- Giroctocogene fitelparvovec was delivered via a single IV infusion to participants in 4 cohorts (n=2 each, except cohort 4: n=5) across 4 ascending doses: 9e11 (cohort 1), 2e12 (cohort 2), 1e13 (cohort 3), and 3e13 vg/kg (cohort 4) (**Figure 2**).
- The high-dose cohort (3e13-vg/kg, cohort 4) was expanded with 3 additional participants; (N=11 across all 4 doses).
- Corticosteroid treatment was initiated for alanine aminotransferase (ALT) elevation that exceeded 1.5× baseline value.
- Safety and efficacy data of each cohort were reviewed by an independent safety monitoring committee prior to each dose escalation and prior to initiating cohort 4 expansion.
- Study endpoints are presented in **Table 1**.

Table 1. Study Endpoints

Primary Endpoints	Secondary Endpoints
• Incidence of AEs and SAEs • Change in circulating FVIII activity	• Change from baseline in the use of FVIII replacement therapy • Change in frequency and severity of bleeding episodes • Measurement of FVIII inhibitor levels • Vector shedding in bodily fluids

AE=adverse event; FVIII=factor VIII; SAE=serious adverse event

RESULTS

- As of the cutoff date (September 6, 2022), participants in all cohorts had been followed for 153 to 263 weeks; all participants have completed ≥35 months (Study Visit: Week 156).
- Participant demographics at baseline are shown in **Table 2**.

Table 2. Participant Demographics by Giroctocogene Fitelparvovec Dose Cohort

Characteristic		Cohort 1 9e11 vg/kg	Cohort 2 2e12 vg/kg	Cohort 3 1e13 vg/kg	Cohort 4 3e13 vg/kg	All Participants
Age, years	n	2	2	2	5	11
	Mean (SD)	30.5 (9.2)	35.5 (16.3)	32.5 (0.7)	27.2 (6.1)	30.3 (7.8)
	Median	30.5	35.5	32.5	29.0	31.0
	Min, max	24, 37	24, 47	32, 33	19, 34	19, 47
Sex, n (%)	Male	2 (100)	2 (100)	2 (100)	5 (100)	11 (100)
Race, n (%)	Asian	–	1 (50)	–	–	1 (9)
	White	2 (100)	1 (50)	2 (100)	4 (80)	9 (82)
	Other	–	–	–	1 (20)	1 (9)
Ethnicity, n (%)	Hispanic or Latino	–	–	–	2 (40)	2 (18)
	Not Hispanic or Latino	2 (100)	2 (100)	2 (100)	3 (60)	9 (82)

Min, max=minimum, maximum; vg=vector genomes

Safety

- Overall, there were 27 treatment-related adverse events (AEs) in 6 participants in cohorts 2 and 4 (**Table 3**). In cohort 4, there were 22 AEs in 4 of 5 participants.

Table 3. Treatment-related Adverse Events by Giroctocogene Fitelparvovec Dose Cohort

MedDRA Preferred Term	Cohort 2: 2e12 vg/kg (n=2) n	Cohort 2: No. of Events	Cohort 4: 3e13 vg/kg (n=5) n	Cohort 4: No. of Events	All Participants (N=11) n	All Participants: No. of Events
Any treatment-related event	2	5	4	22	6	27
Grade 3/4 AE	0	0	1[a]	1	1	1
ALT increased	2	3	3	10	5	13
AST increased	1	2	2	3	3	5
Pyrexia	0	0	3	3	3	3
Tachycardia	0	0	2	2	2	2
Myalgia	0	0	1	1	1	1
Hypotension	0	0	1	1	1	1
Fatigue	0	0	1	1	1	1
FVIII level increased	0	0	1	1	1	1

[a] One participant experienced grade 3 hypotension that was considered related to study drug and resolved with treatment.
AE=adverse event; ALT=alanine transaminase; AST=aspartate aminotransferase; FVIII=coagulation factor VIII; MedDRA=Medical Dictionary for Regulatory Activities; vg=vector genomes

- Treatment-related serious AEs were reported in 1 participant (in cohort 4) who experienced hypotension (grade 3) and fever (grade 2) with onset ~6 h after giroctocogene fitelparvovec infusion.
 - The events fully resolved with treatment and did not delay post-infusion discharge from the [clinic/hospital/etc] the next day.
 - Following implementation of additional supportive care measures, there were no similar occurrences of hypotension.
- AEs (all-causality) of ALT increases requiring ≥7 days of corticosteroids were observed in 4 of the 5 participants in cohort 4; these AEs were classified as treatment-related in 3 participants.
 - Elevations in ALT were managed with a tapering course of corticosteroids: median duration: 56 days (range, 7–135 days).
 - Efficacious levels of FVIII activity were maintained.
- There were no confirmed instances of inhibitor to FVIII across all of the study participants (N=11).
- There have been no thrombotic events or liver masses detected.

Efficacy (cohort 4)

- FVIII activity levels are shown in **Figure 3** and **Table 4**.
- All 5 participants had data available at Week 156.
 - Four participants had FVIII in the mild to normal range.
 - One participant (#02) had FVIII activity levels below the lower level of quantification (<3%), as measured with a chromogenic assay, and 3.3% measured with a 1-stage assay.
- Two participants had data available through Week 182. Both maintained FVIII in the mild to normal range.

Figure 3. Factor VIII Activity Levels (Measured With Chromogenic Assay) for Individuals in the Giroctocogene Fitelparvovec Highest Dose Cohort (3e13 vg/kg, Cohort 4)

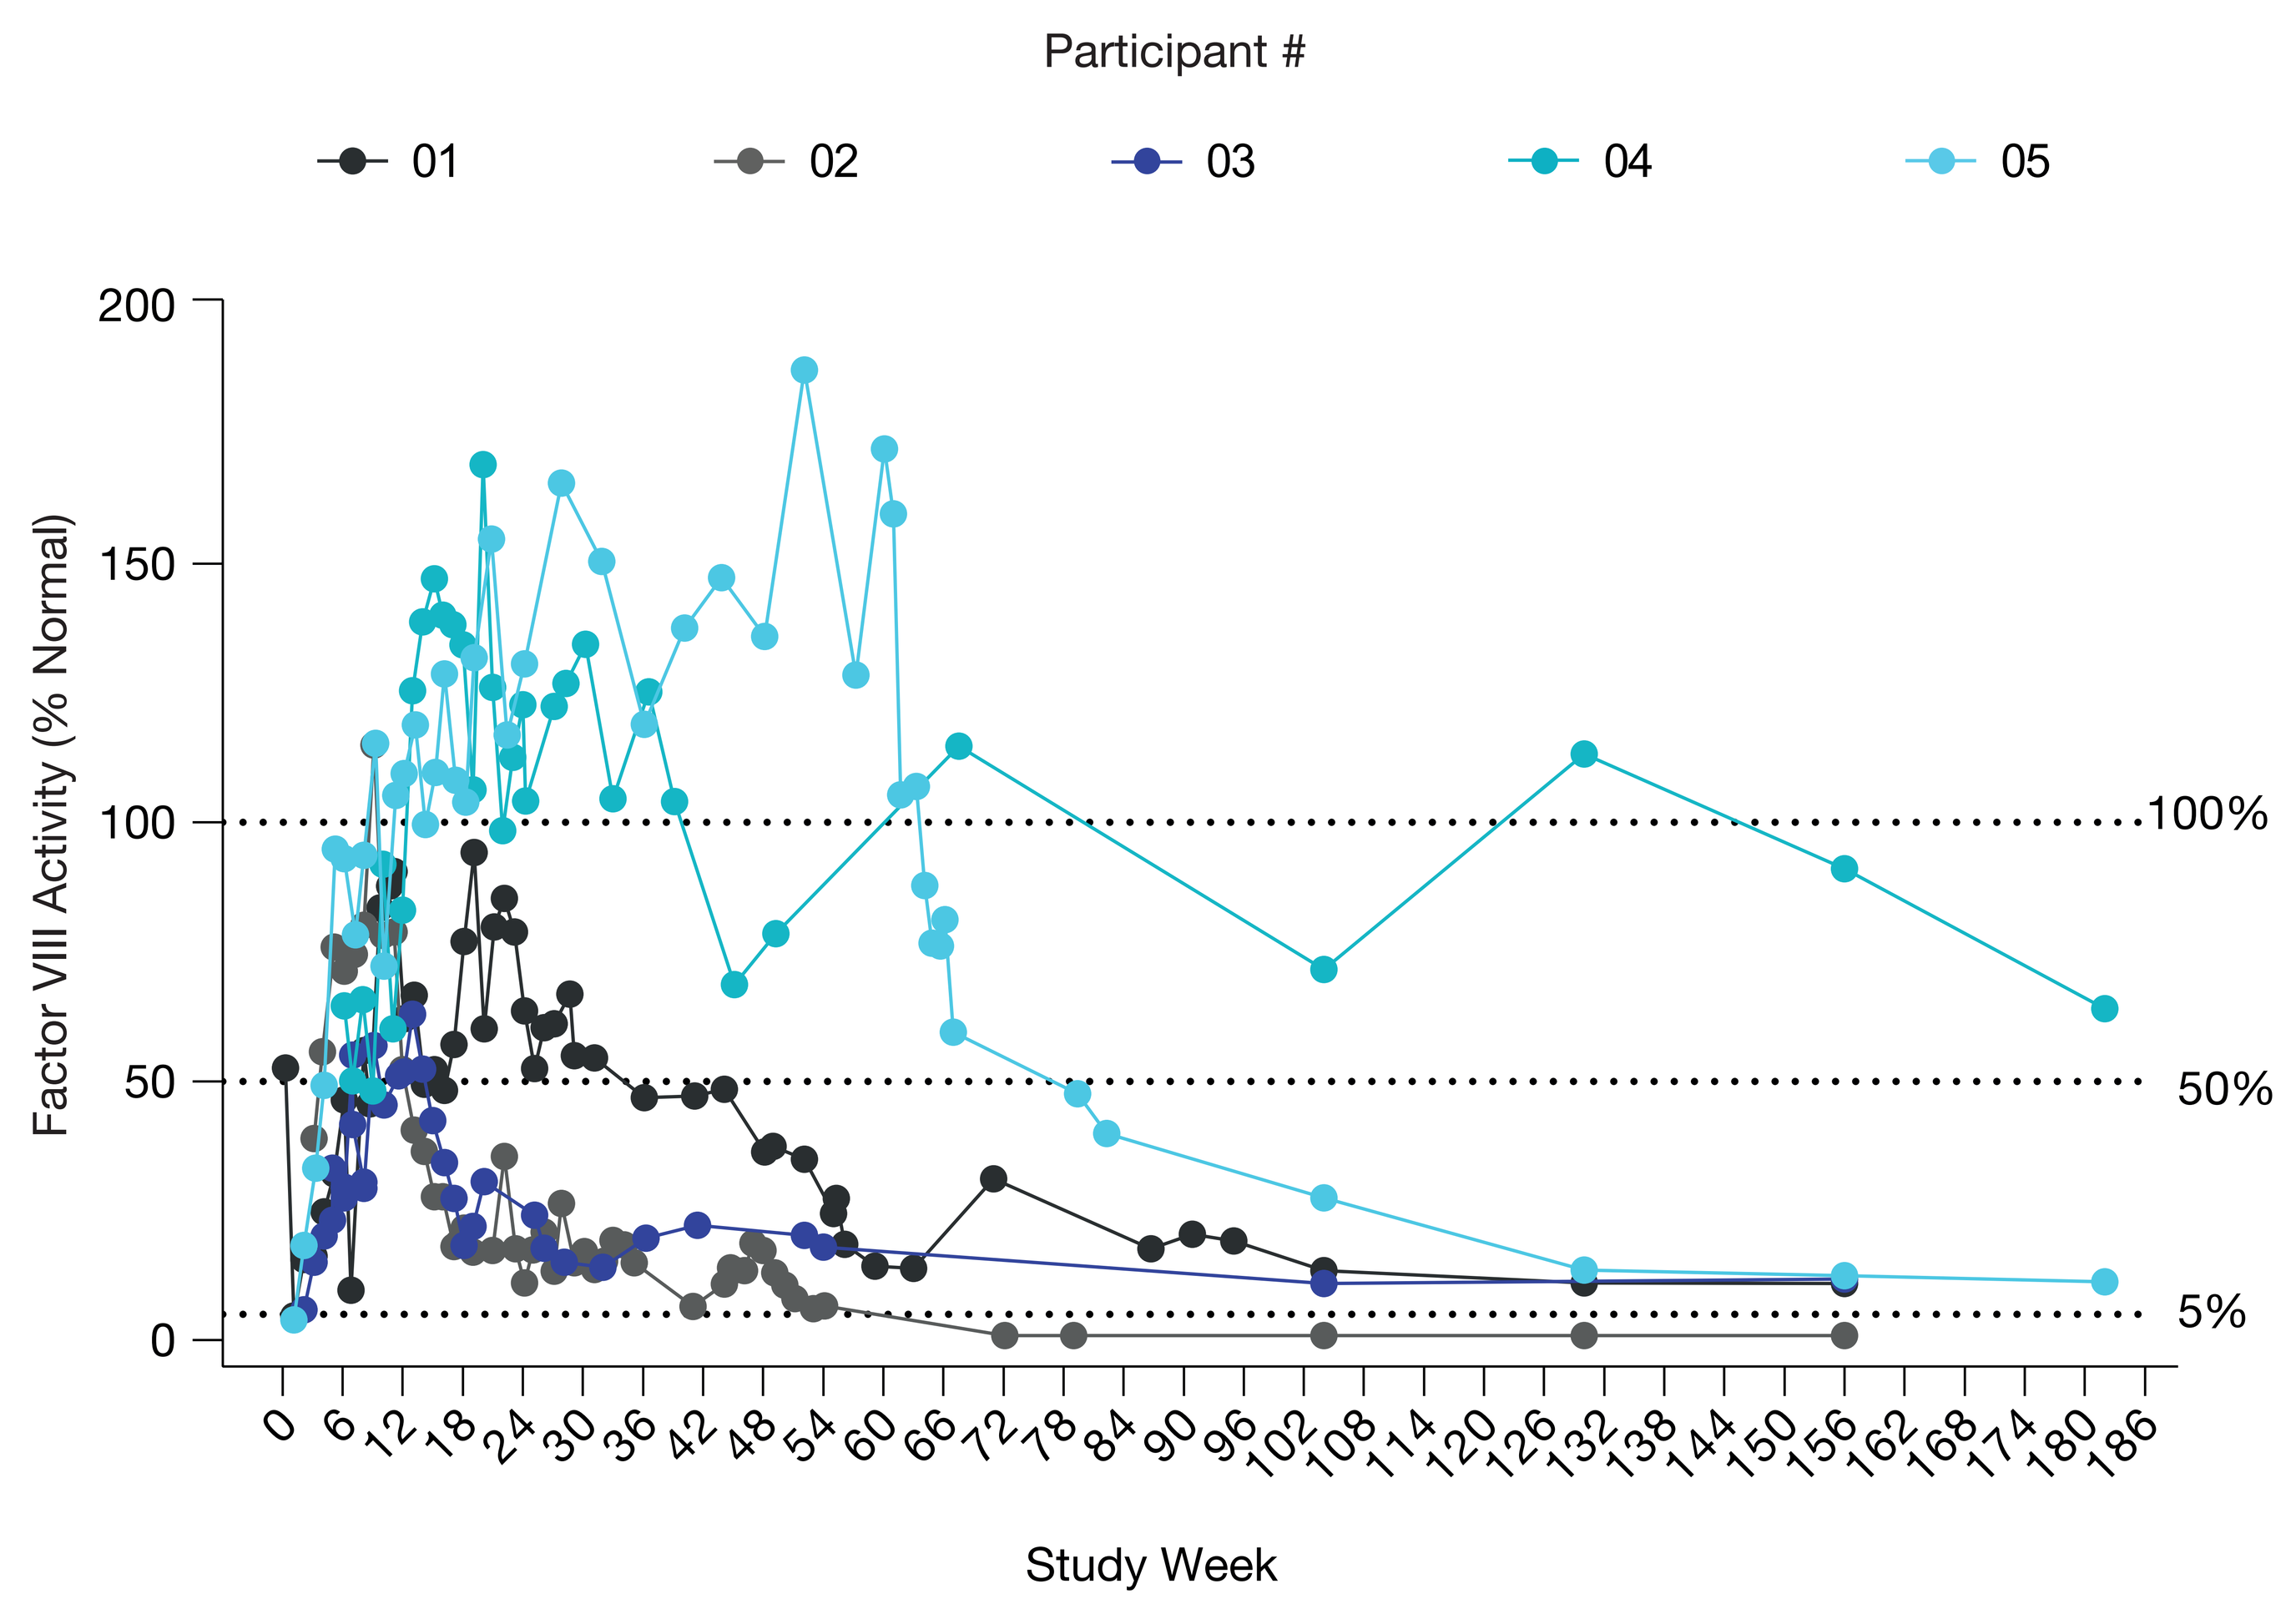
Participant #
01
02
03
04
05
Factor VIII Activity (% Normal)
0
50
100
150
200
100%
50%
5%
Study Week
0 6 12 18 24 30 36 42 48 54 60 66 72 78 84 90 96 102 108 114 120 126 132 138 144 150 156 162 168 174 180 186

FVIII activity values collected within 96 h of an FVIII replacement therapy infusion were excluded. For results reported as below the limit of quantification (<3%), a value of 0.009 IU/mL (0.9%) was used for analysis and plotting.
Mild range: >5% to <40%; normal range: >50%; FVIII=coagulation factor VIII; vg=vector genomes

Table 4. Mean Factor VIII Activity Level by 1-Stage and Chromogenic Assay for the Giroctocogene Fitelparvovec Highest Dose Cohort (3e13-vg/kg, Cohort 4)

Assay	Week 12	Week 24	Week 52	Week 78	Week 104	Week 130	Week 156	Week 182
	Factor VIII Activity, % Normal, Mean (min, max)							
Participants, n	5	5	4[a]	4[a]	5	4[a]	5	2[b]
1-stage clotting	110.9 (82.7, 167.7)	107.5 (30.5, 212.6)	66.4 (12.0, 191.3)	65.7 (3.8, 144.2)	38.9 (4.1, 99.1)	54.1 (5.4, 164.5)	40.5 (3.3, 129.0)	66.0 (18.2, 113.9)
Chromogenic	71.7 (51.8, 109.5)	68.9 (20.4, 123.8)	42.6 (7.8, 122.3)	48.9 (BLQ, 114.7)	25.4 (BLQ, 71.6)	34.7 (BLQ, 113.2)	25.5 (BLQ, 91.1)	37.7 (11.3, 64.0)

[a] There was 1 participant each who was unable to attend visits at Weeks 52, 78, and 130.
[b] Three participants had not yet reached Week 182 at the time of the data cutoff.
BLQ=below limit of quantification (<1% for 1-Stage Assay and <3% for Chromogenic Assay); min, max=minimum, maximum

- The mean annualized total bleeding rate [(number of all bleeding episodes starting 3 weeks after study drug infusion) / (observation period in years)] was 0 for the first-year post-infusion and 1.2 throughout the total duration of follow-up.
 - 2 participants experienced an overall total of 18 bleeding events:
 - 1 participant (#03) (1 event): circumstances of bleed unknown; event occurred in a prior target joint on Study Day 471 (~Week 67).
 - 1 participant (#02) (17 events): 8 traumatic, 5 spontaneous, 4 unknown; no events occurred in known prior target joints; first occurrence on Study Day 474 (~Week 68).
- No participants in cohort 4 have resumed prophylaxis.

CONCLUSIONS

- A single infusion of giroctocogene fitelparvovec gene therapy in participants with severe hemophilia A remains generally well tolerated over a period of 3 years post-infusion. There was:
 - an increase in FVIII levels into the moderate to normal range for participants in the highest-dose cohort (3e13 vg/kg, cohort 4).
 - no sustained AEs.
 - minimal overall bleeding in highest-dose cohort (cohort 4).
- The ongoing phase 3 study (NCT04370054) in a larger cohort will provide more long-term data to further characterize the safety and durability of giroctocogene fitelparvovec in participants with moderately severe to severe hemophilia A.

REFERENCES

1. Srivastava A, et al. Haemophilia 2020;26(suppl 6):1-158.
2. White GC, et al. Thromb Haemost 2001;85:560.
3. Leebeek FWG, et al. Blood 2021;138:923-931.

ACKNOWLEDGMENTS

This study was funded by Pfizer. Medical writing support was provided by Courtney Cameron, PhD, of Engage Scientific Solutions and funded by Pfizer.
Copyright © 2022

Please scan this QR code with your smartphone app to view or obtain a copy of this poster. If you don't have a smartphone, access the poster via the internet at: https://scientificpubs.congressposter.com/p/usaan717o3ny1f0c

Presented at the American Society of Hematology (ASH) Annual Meeting 2022, December 10–13, 2022, New Orleans, LA, USA